IDWeek 2026 | Poster #P-168

Activity of Cefiderocol-Xeruborbactam and Comparator Agents against *Pseudomonas aeruginosa*, Including Molecularly Characterized Isolates Recovered Globally in 2024

KM Papp-Wallace[1], JH Kimbrough[1], JM Maher[1], RE Mendes[1], HS Sader[1], and M Castanheira[1]

[1]Element Iowa City (JMI Laboratories), North Liberty, Iowa, USA

Background

- Cefiderocol-xeruborbactam is a combination of a siderophore-conjugated cephalosporin with a boronate β-lactamase inhibitor and this combination has notable antimicrobial activity against Gram-negative pathogens.
- Herein, the activity of cefiderocol-xeruborbactam and comparators was evaluated against *P. aeruginosa* collected from the United States, Europe, Turkey, and Israel in 2024.

Methods

- A total of 2,185 *P. aeruginosa* were consecutively collected from 69 hospitals in 20 countries through the SENTRY Surveillance Program.
- Susceptibility testing was performed using broth microdilution with cation-adjusted Mueller-Hinton broth (CAMHB) for all agents except cefiderocol, which was tested in iron-depleted CAMHB.
- CLSI breakpoints were applied. *P. aeruginosa* isolates displaying MIC values of ≥4 mg/L for meropenem or imipenem were screened for carbapenemases.

Results

- Cefiderocol-xeruborbactam exhibited potent activity against *P. aeruginosa* collected from the US (Figure 1) and Europe (Figure 2) with 99.6% of isolates with an MIC ≤4 mg/L and $MIC_{50/90}$ values of ≤0.06/0.25 mg/L, while % susceptibility varied from 91.4–99.9% for comparators (Table 1).
- Besides colistin, cefiderocol-xeruborbactam was the most active antibiotic tested with 92.9–99.2% of isolates having an MIC ≤4 mg/L including multidrug-resistant (MDR), extensively drug-resistant (XDR), ceftolozane-tazobactam-non-susceptible (NS), and carbapenem-NS with and without carbapenemases (Table 1 and Figure 3).
- Against MDR isolates, comparators, except for meropenem-vaborbactam, were effective with 71.8–100% of isolates testing susceptible. XDR isolates were not as susceptible to most comparators (29.4–49.7%) except for cefiderocol at 92.2% and colistin at 100% (Table 1 and Figure 3).
- *P. aeruginosa* from Europe were less susceptible to cefiderocol-xeruborbactam, cefiderocol, and ceftazidime-avibactam compared to isolates from the US (Figure 3).
- European isolates were more susceptible to ceftolozane-tazobactam, imipenem-relebactam, and meropenem-vaborbactam compared to US isolates (Figure 3).
- Carbapenemase positive *P. aeruginosa* were mostly collected from Europe, while a single isolate with a carbapenemase was identified in US isolates (Figure 3).
- Results for isolates with or without carbapenemases were mixed with cefiderocol-xeruborbactam performing well; 92.9–99.2% of isolates had an MIC ≤4 mg/L (Table 1 and Figure 3).
- Ceftolozane-tazobactam, imipenem-relebactam, meropenem-vaborbactam, and ceftazidime-avibactam showed 2.4–4.8% susceptibility against carbapenemase-producers while cefiderocol was at 76.2% (Table 1 and Figure 3).
- Activity was better against non-carbapenemase producers with ceftolozane-tazobactam, imipenem-relebactam, meropenem-vaborbactam, and ceftazidime-avibactam and cefiderocol displaying 69.8–99.2% susceptibility (Table 1 and Figure 3).

Conclusions

- Cefiderocol-xeruborbactam was very active against *P. aeruginosa*, including isolates producing carbapenemases or non-susceptible to ceftolozane-tazobactam.
- These data support further development of cefiderocol-xeruborbactam for the treatment of infections caused by *P. aeruginosa*.

Acknowledgments

This study at Element Iowa City/JMI Laboratories was supported by Qpex Biopharma (San Diego, CA). Element Iowa City/JMI Laboratories received compensation fees for services in relation to preparing the poster, which was funded by Qpex. This project has been funded in whole or in part with federal funds from the Department of Health and Human Services; Administration for Strategic Preparedness and Response; Center for the Biomedical Advanced Research and Development Authority (BARDA), under OTA number HHSO100201600026C.

References

CLSI. 2026. M100Ed36. Performance standards for antimicrobial susceptibility testing: 36th informational supplement. Clinical and Laboratory Standards Institute.

Contact

Krisztina M. Papp-Wallace, PhD
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Email: KrisztinaPappWallace@element.com

Scan Me

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2026_26-QPX-02_P2_CFDC-Xeru_PSA.pdf

Charges may apply. No personal information is stored.

Figure 1. U.S. Census division distribution of *P. aeruginosa* and cefiderocol-xeruborbactam AST results (n=1,078)

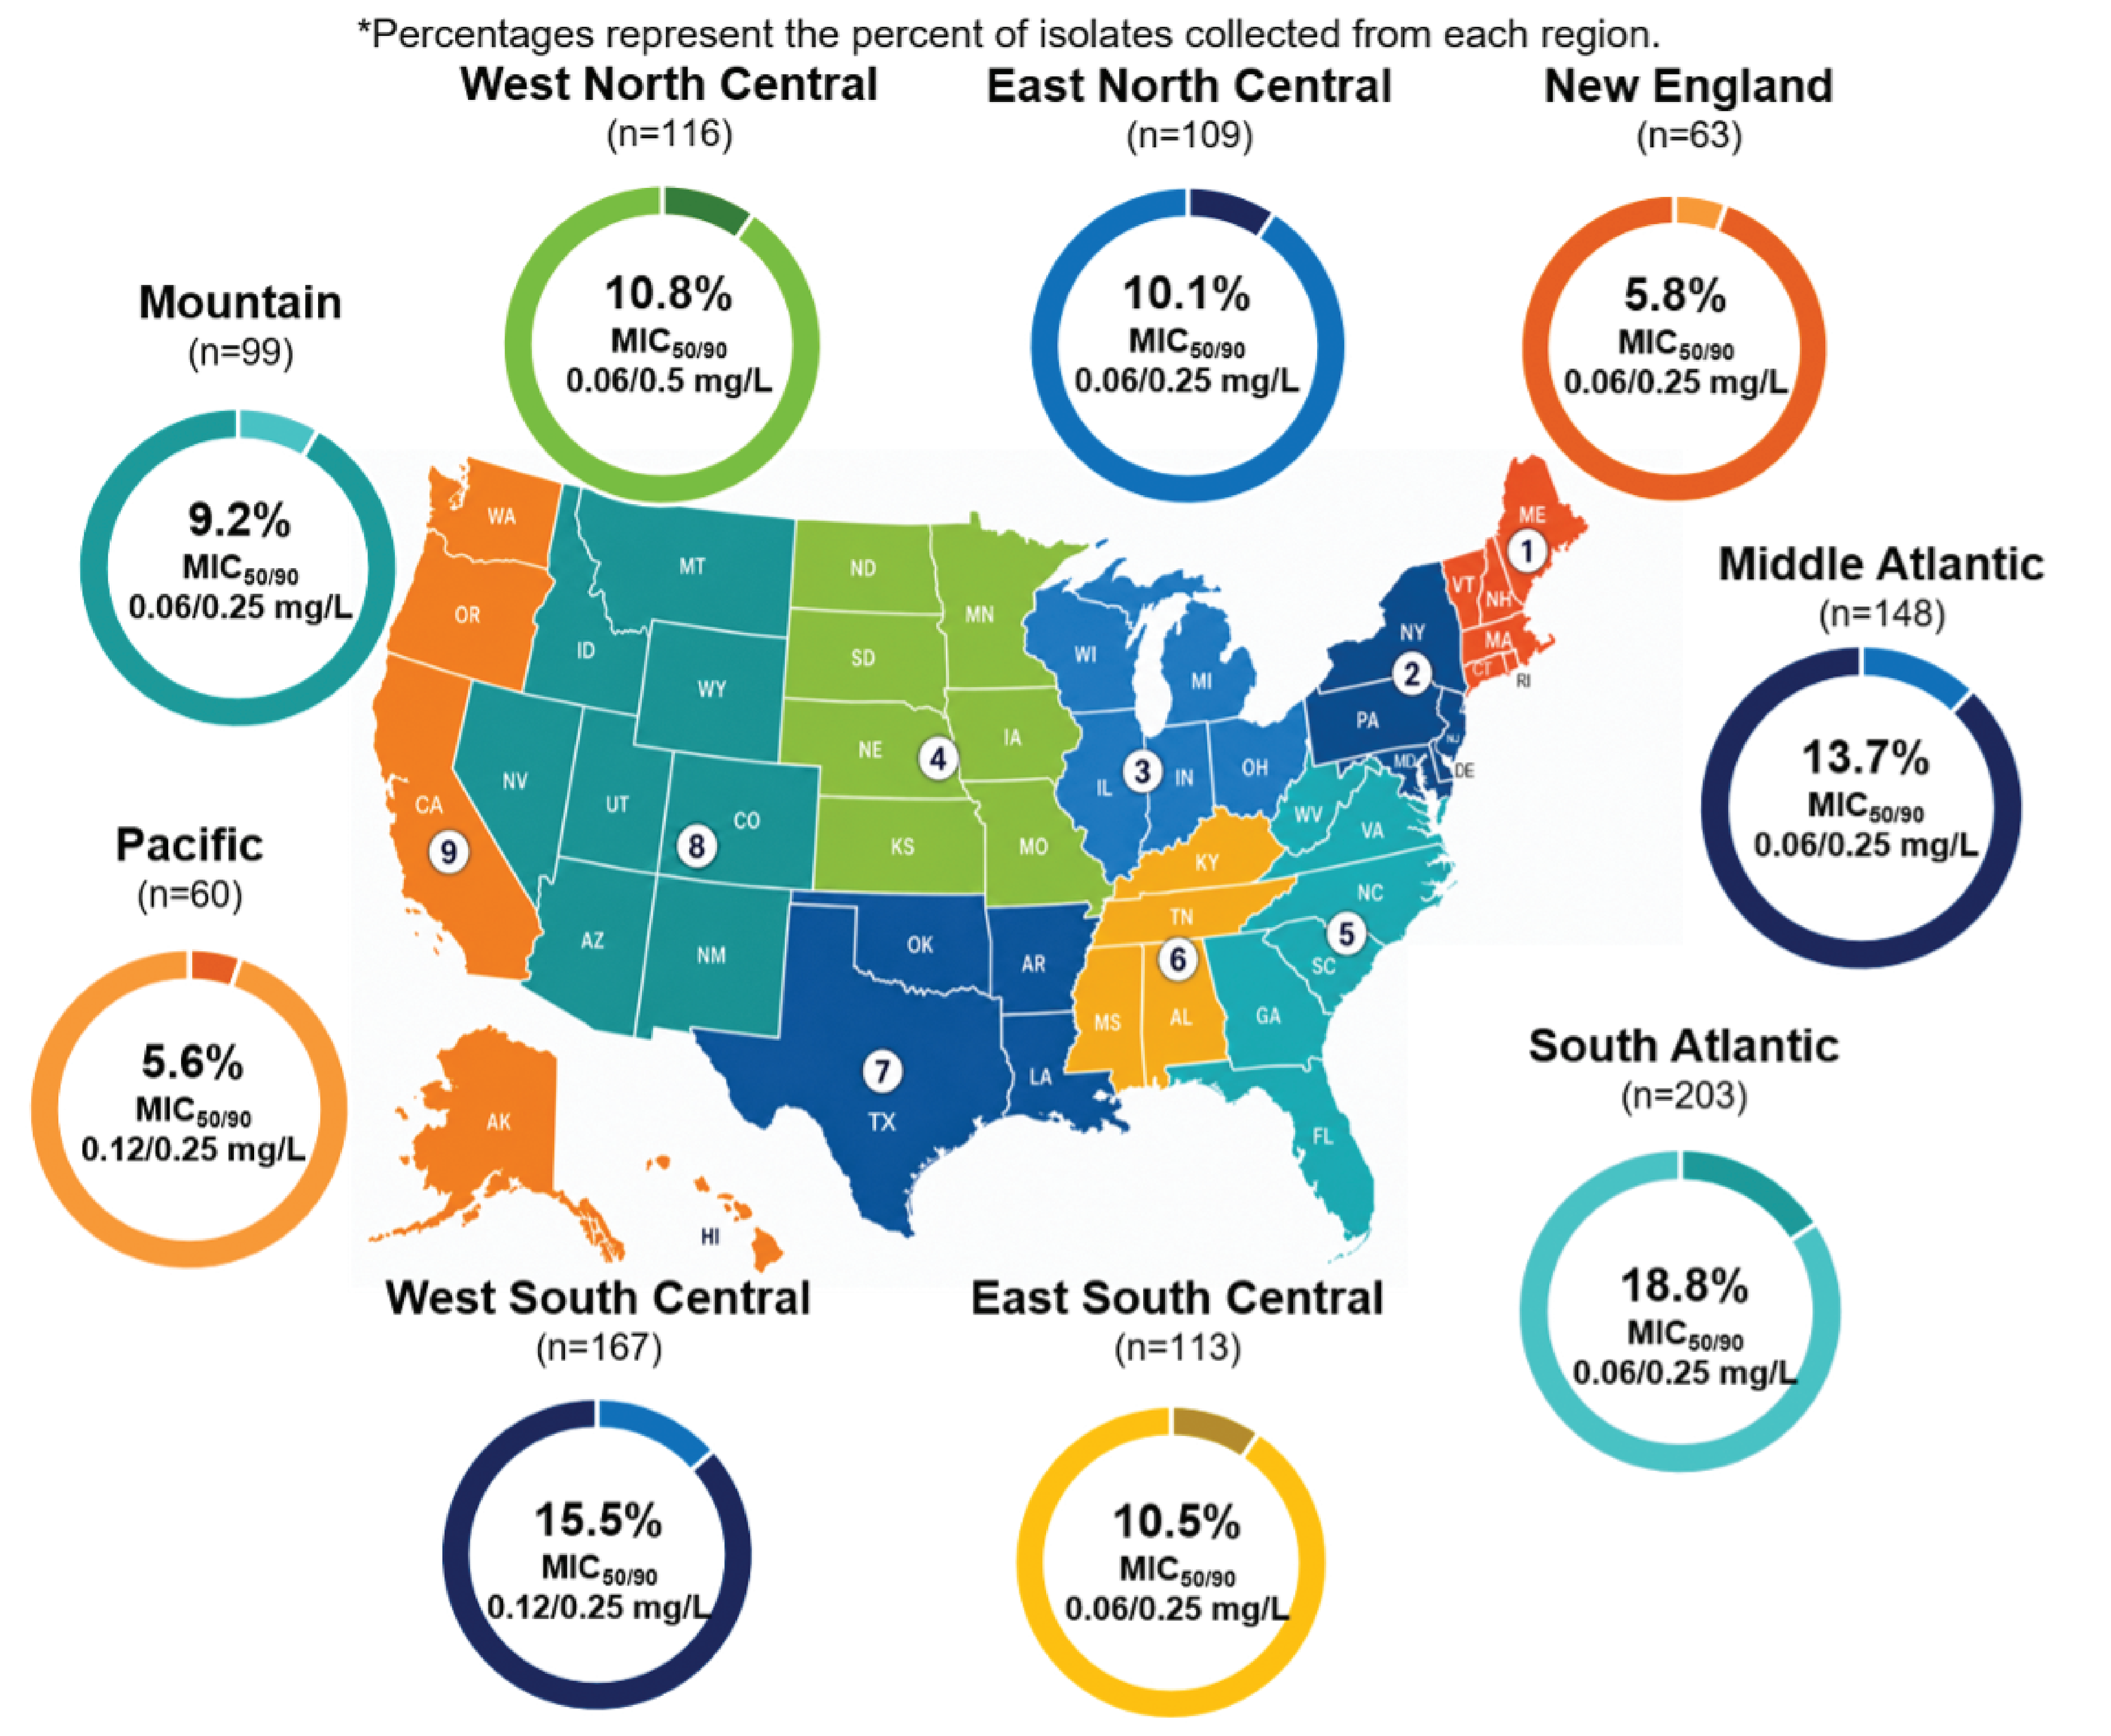
*Percentages represent the percent of isolates collected from each region.
West North Central (n=116) 10.8% MIC50/90 0.06/0.5 mg/L
East North Central (n=109) 10.1% MIC50/90 0.06/0.25 mg/L
New England (n=63) 5.8% MIC50/90 0.06/0.25 mg/L
Mountain (n=99) 9.2% MIC50/90 0.06/0.25 mg/L
Middle Atlantic (n=148) 13.7% MIC50/90 0.06/0.25 mg/L
Pacific (n=60) 5.6% MIC50/90 0.12/0.25 mg/L
South Atlantic (n=203) 18.8% MIC50/90 0.06/0.25 mg/L
West South Central (n=167) 15.5% MIC50/90 0.12/0.25 mg/L
East South Central (n=113) 10.5% MIC50/90 0.06/0.25 mg/L

Figure 2. European distribution of *P. aeruginosa* and cefiderocol-xeruborbactam AST results (n=1,107)

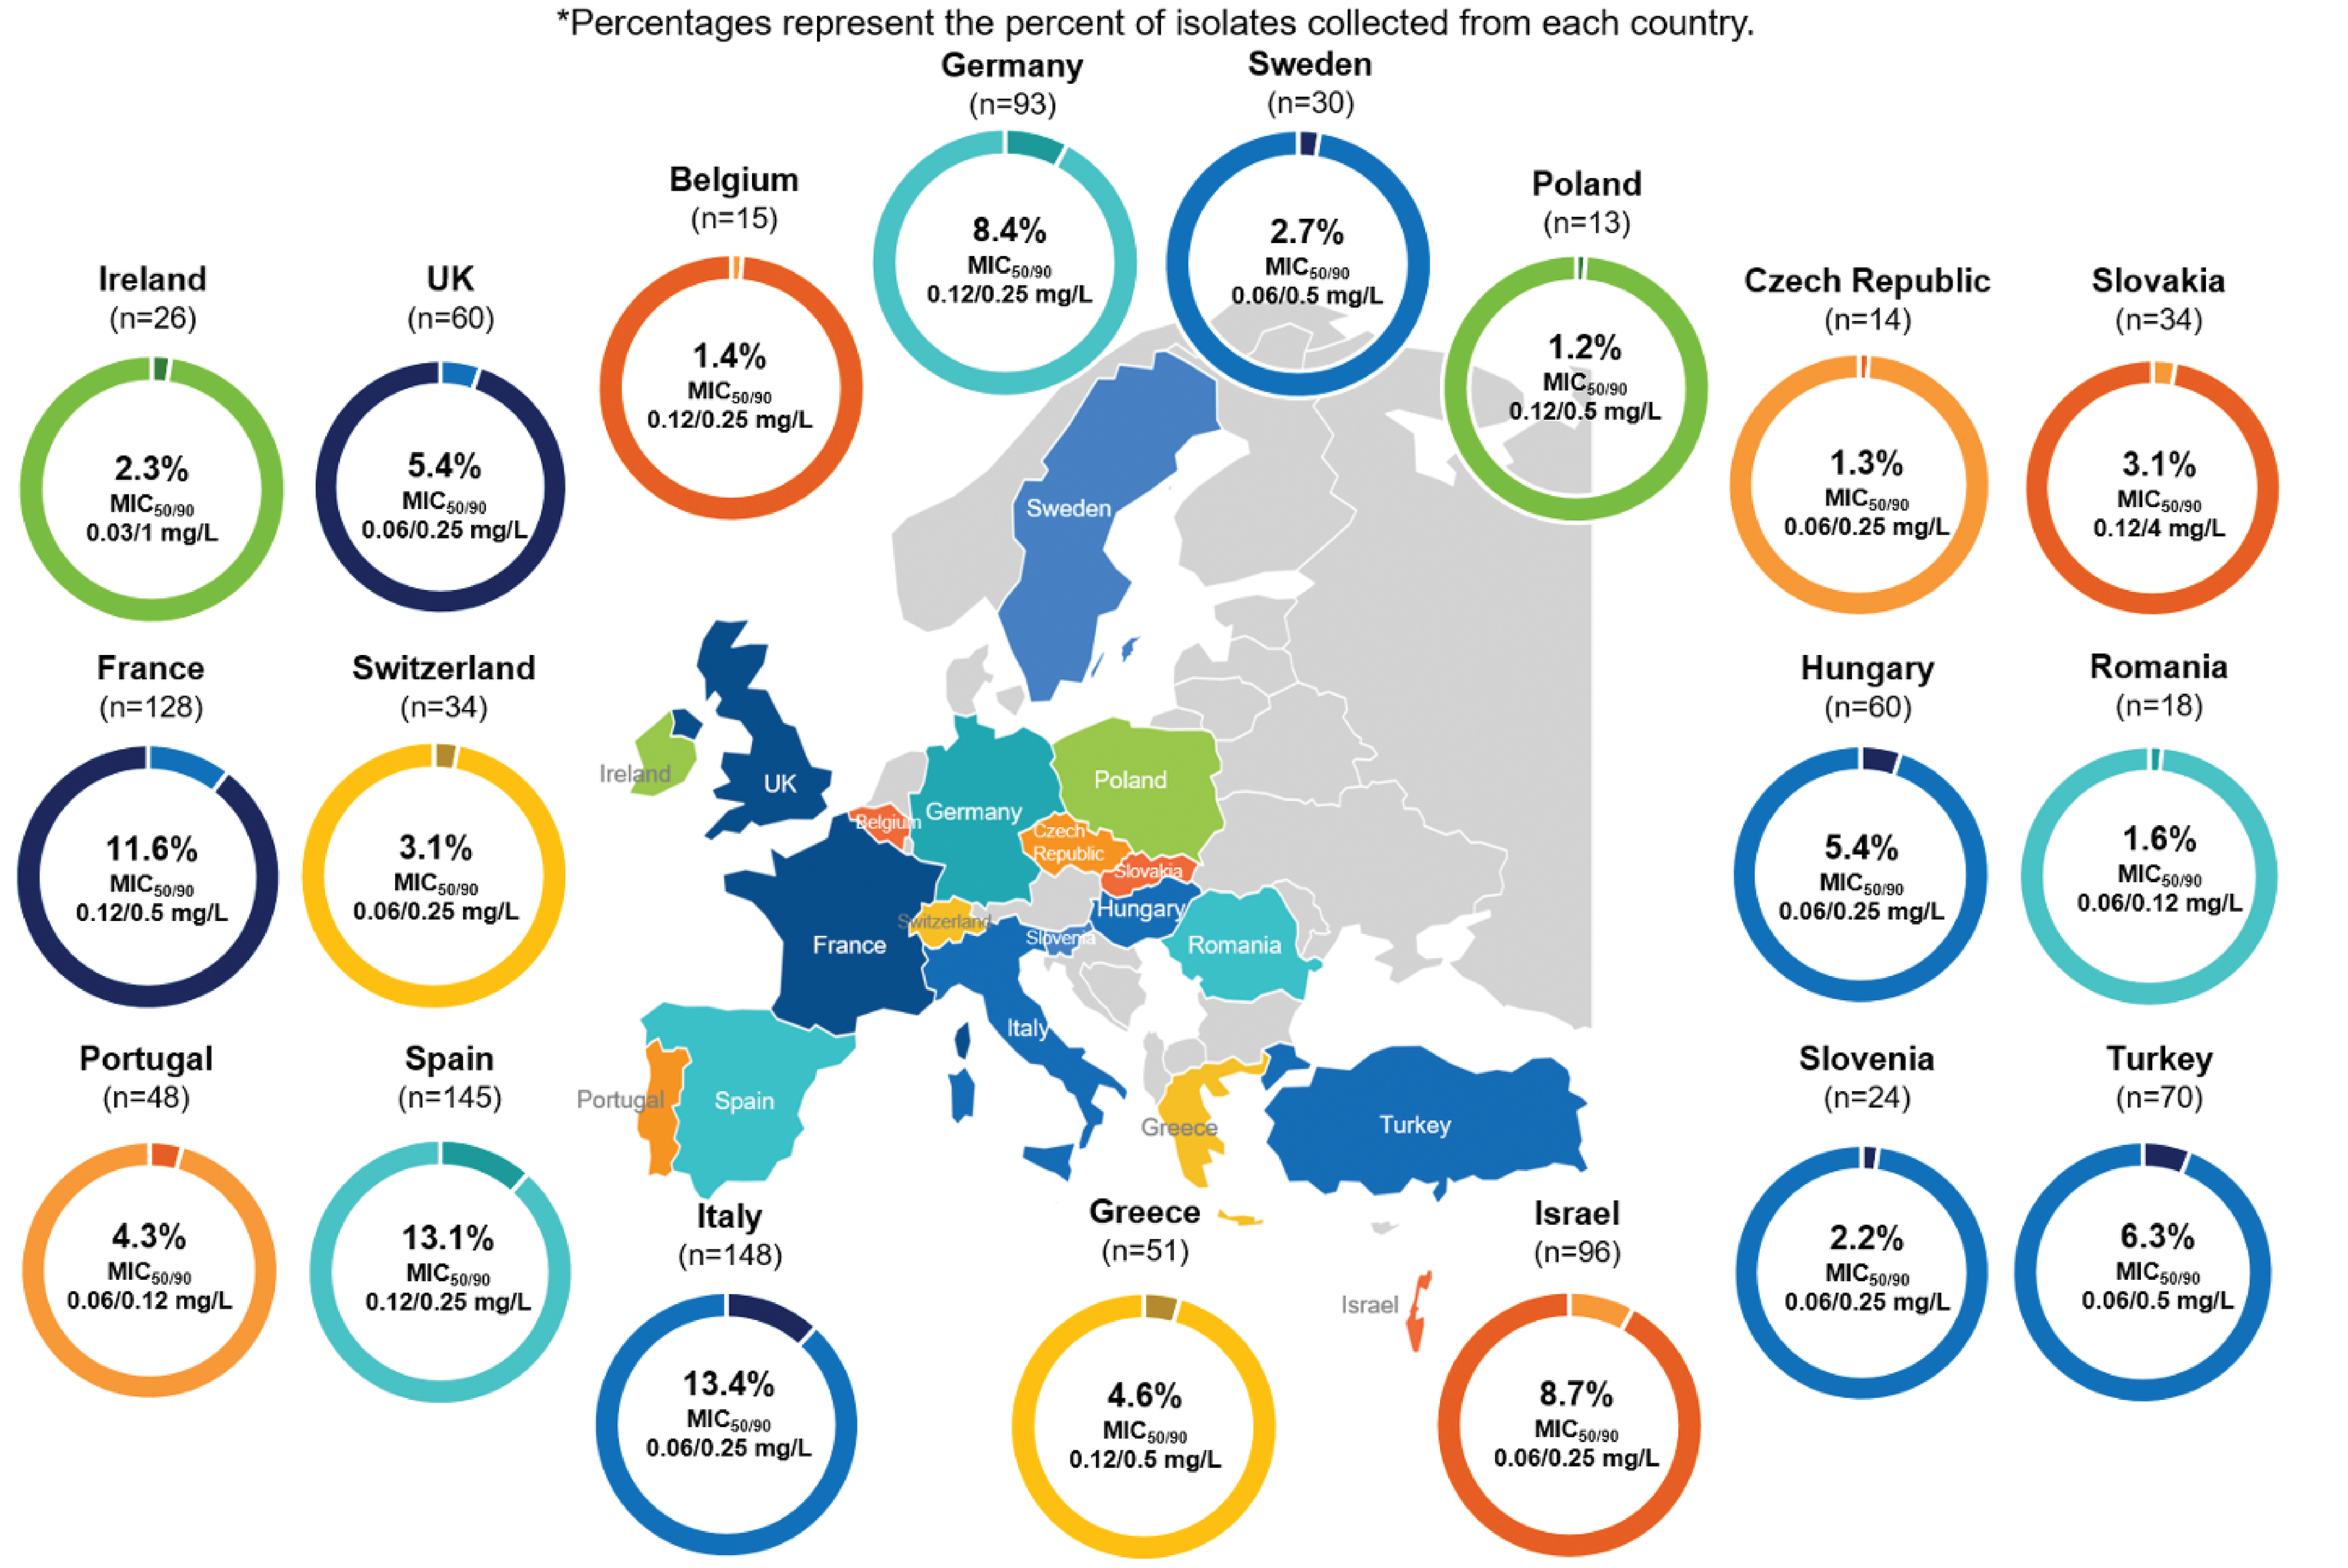
*Percentages represent the percent of isolates collected from each country.
Germany (n=93) 8.4% MIC50/90 0.12/0.25 mg/L
Sweden (n=30) 2.7% MIC50/90 0.06/0.5 mg/L
Belgium (n=15) 1.4% MIC50/90 0.12/0.25 mg/L
Poland (n=13) 1.2% MIC50/90 0.12/0.5 mg/L
Ireland (n=26) 2.3% MIC50/90 0.03/1 mg/L
UK (n=60) 5.4% MIC50/90 0.06/0.25 mg/L
Czech Republic (n=14) 1.3% MIC50/90 0.06/0.25 mg/L
Slovakia (n=34) 3.1% MIC50/90 0.12/4 mg/L
France (n=128) 11.6% MIC50/90 0.12/0.5 mg/L
Switzerland (n=34) 3.1% MIC50/90 0.06/0.25 mg/L
Hungary (n=60) 5.4% MIC50/90 0.06/0.25 mg/L
Romania (n=18) 1.6% MIC50/90 0.06/0.12 mg/L
Portugal (n=48) 4.3% MIC50/90 0.06/0.12 mg/L
Spain (n=145) 13.1% MIC50/90 0.12/0.25 mg/L
Slovenia (n=24) 2.2% MIC50/90 0.06/0.25 mg/L
Turkey (n=70) 6.3% MIC50/90 0.06/0.5 mg/L
Italy (n=148) 13.4% MIC50/90 0.06/0.25 mg/L
Greece (n=51) 4.6% MIC50/90 0.12/0.5 mg/L
Israel (n=96) 8.7% MIC50/90 0.06/0.25 mg/L

Figure 3. Antimicrobial activity of cefiderocol-xeruborbactam and comparators against Enterobacterales stratified by phenotype and region

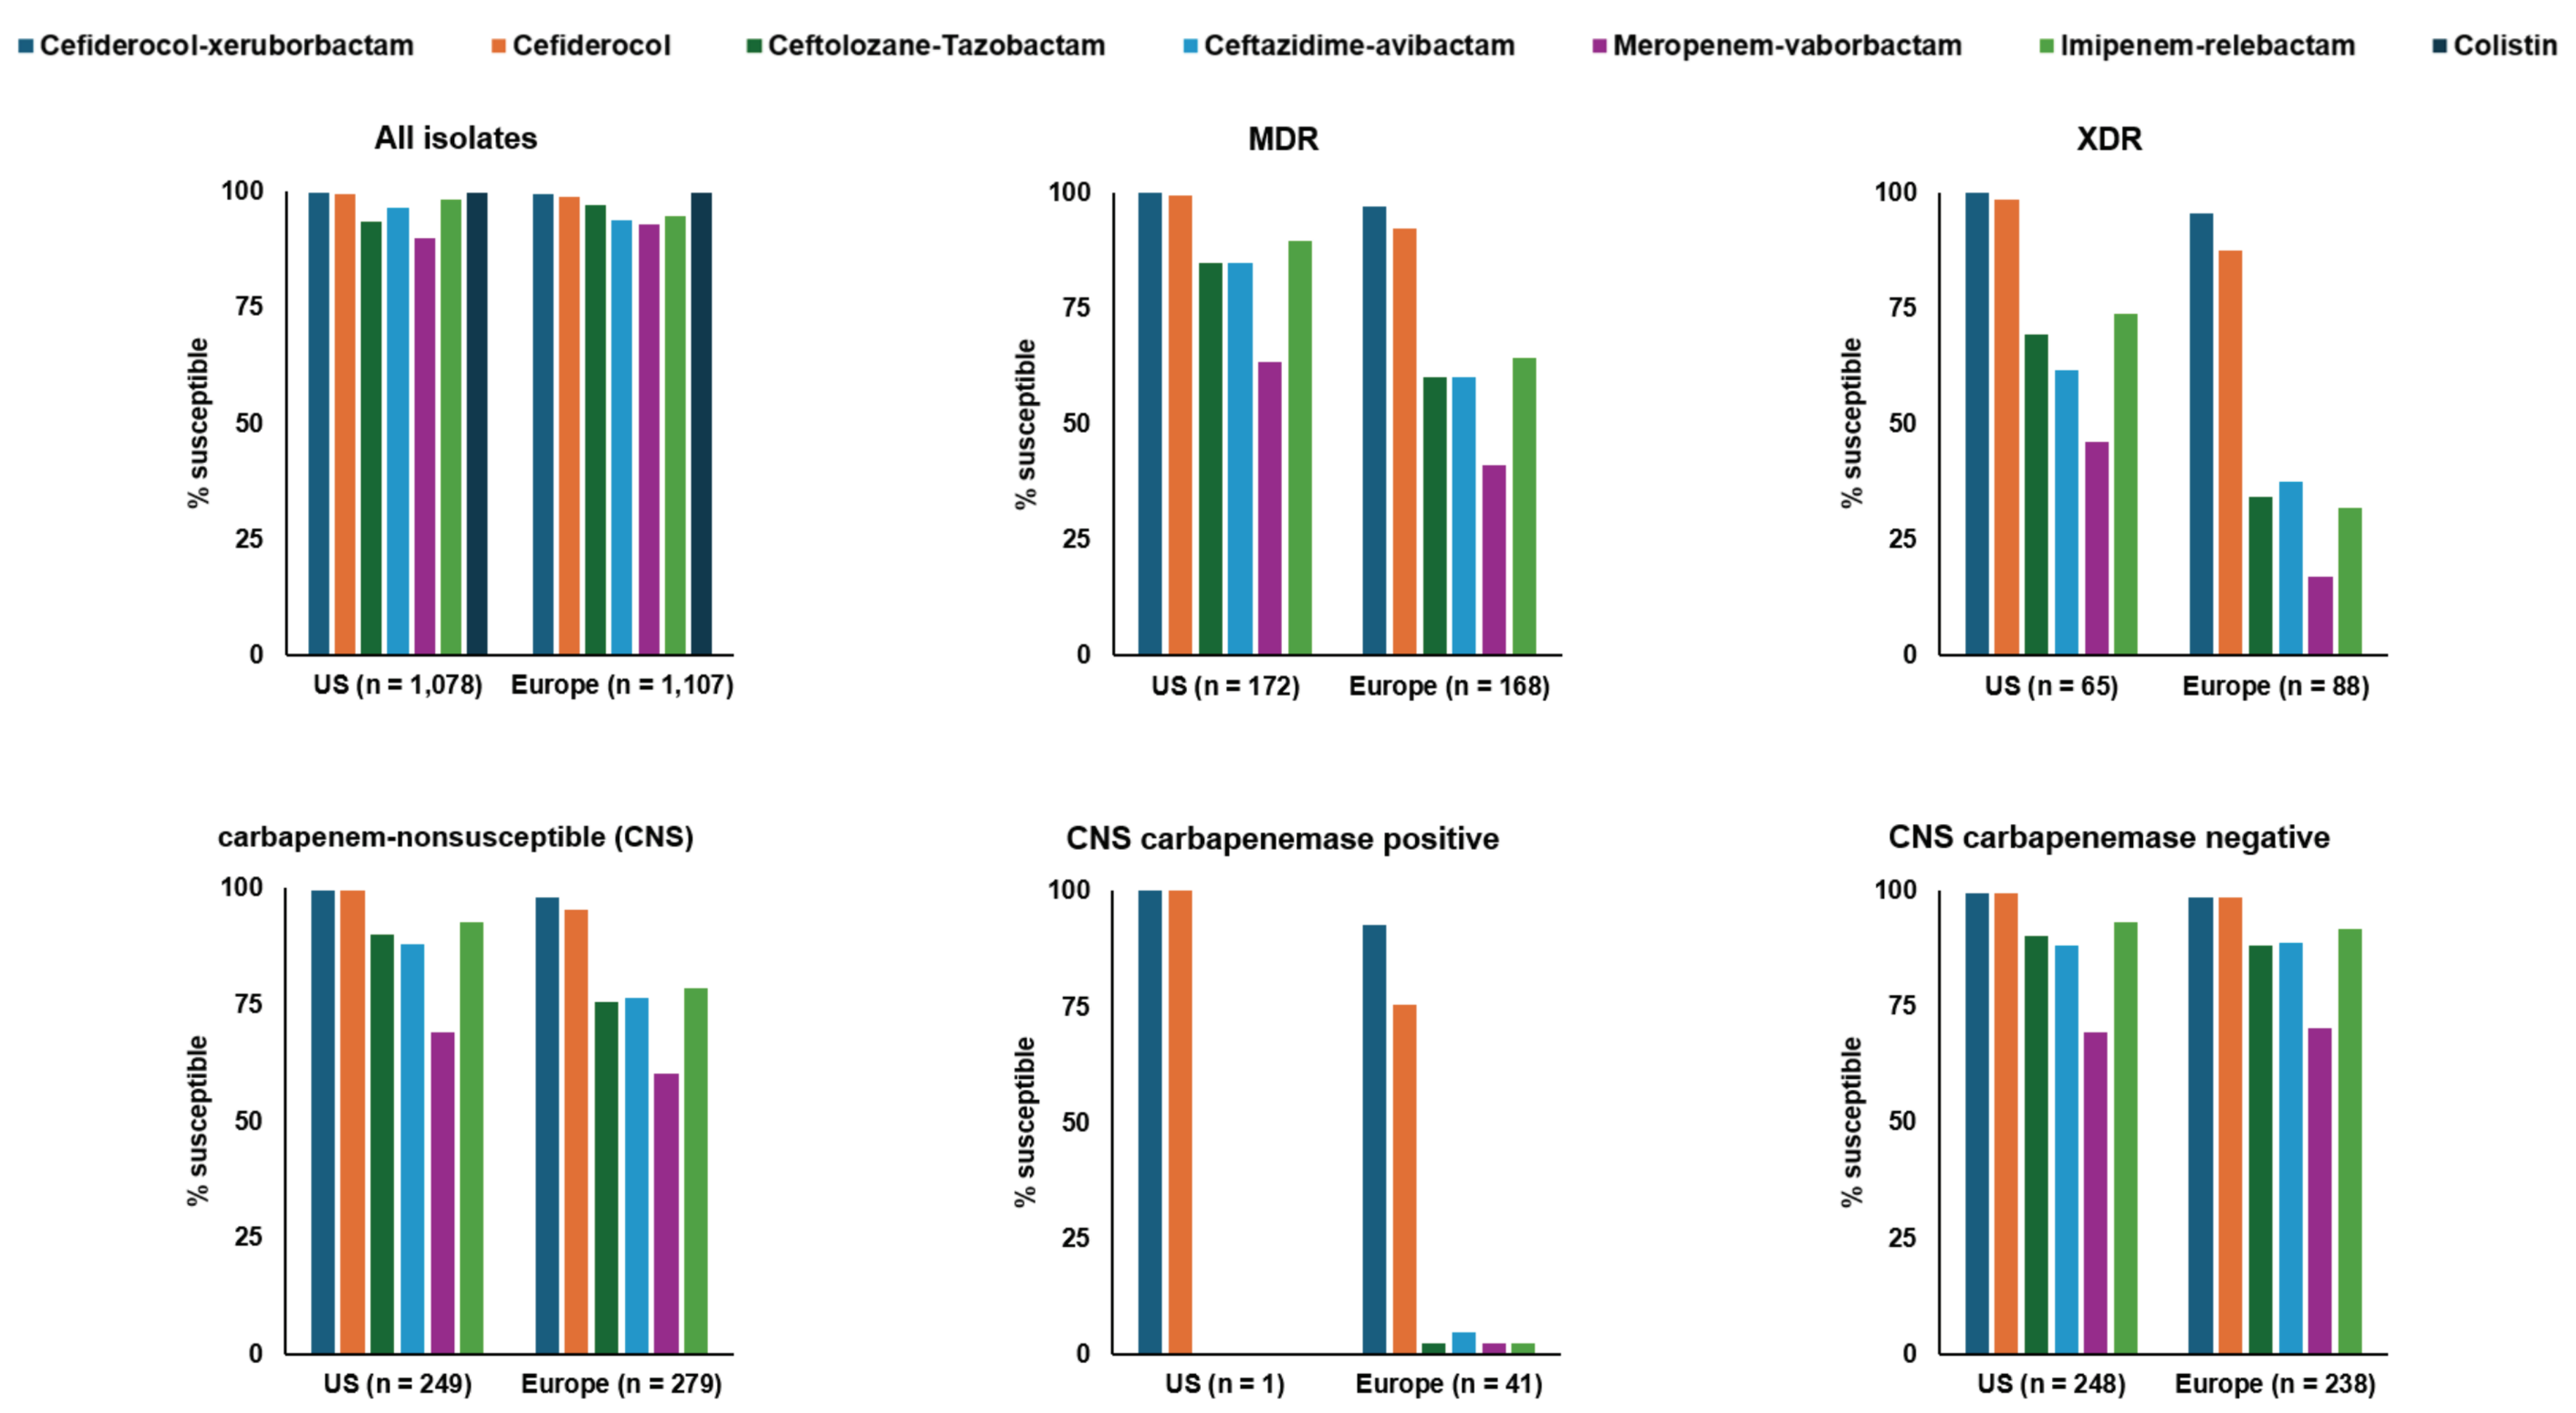
Cefiderocol-xeruborbactam
Cefiderocol
Ceftolozane-Tazobactam
Ceftazidime-avibactam
Meropenem-vaborbactam
Imipenem-relebactam
Colistin
All isolates
US (n = 1,078)
Europe (n = 1,107)
MDR
US (n = 172)
Europe (n = 168)
XDR
US (n = 65)
Europe (n = 88)
carbapenem-nonsusceptible (CNS)
US (n = 249)
Europe (n = 279)
CNS carbapenemase positive
US (n = 1)
Europe (n = 41)
CNS carbapenemase negative
US (n = 248)
Europe (n = 238)
% susceptible
0
25
50
75
100

Table 1. Antimicrobial activity of cefiderocol-xeruborbactam and comparators against *P. aeruginosa*

%Susceptible: ≥99% | 98.9-90% | 89.9-60% | 59.9-40% | 39.9-0%

Phenotype/genotype (No.)	MIC_{50}/MIC_{90} in mg/L (% susceptible by CLSI criteria)[a]						
	Cefiderocol-xeruborbactam [b]	Cefiderocol	Ceftolozane-Tazobactam	Imipenem-relebactam	Meropenem-vaborbactam	Ceftazidime-avibactam	Colistin
All (2,185)	≤0.06/0.25 99.6%	0.12/0.5 99.2%	0.5/2 95.3%	0.25/1 96.4%	0.5/8 91.4%	2/8 95.1%	0.5/1 99.9%
MDR (340)[c]	0.12/1 98.5%	0.25/2 95.9%	2/>16 72.6%	1/>16 77.1%	8/>32 52.4%	4/>32 71.8%	0.5/1 100%
XDR (153)[c]	0.12/2 97.4%	0.25/4 92.2%	8/>16 49.0%	4/>16 49.7%	16/>32 29.4%	16/>32 47.7%	0.5/1 100%
Ceftolozane-Tazobactam-NS (102)[d]	0.25/2 96.1%	0.5/4 86.3%	>16/>16 0.0%	4/>16 41.2%	32/>32 25.5%	32/>32 23.5%	0.5/1 100%
Carbapenem-NS (528)[d]	0.12/1 98.7%	0.12/1 97.3%	1/>16 82.4%	1/8 85.2%	8/32 64.4%	4/32 81.8%	0.5/1 100%
Carbapenemase + (42)[e]	1/4 92.9%	2/16 76.2%	>16/>16 2.4%	>16/>16 2.4%	>32/>32 2.4%	>32/>32 4.8%	1/1 100%
Carbapenemase – (486)	0.06/0.5 99.2%	0.12/0.5 99.2%	1/8 89.3%	1/2 92.4%	4/16 69.8%	4/16 88.3%	0.5/1 100%

Abbreviations: C/T, ceftolozane-tazobactam; MDR, multidrug-resistant; XDR, extensively drug-resistant; NS, non-susceptible; +, positive; -, negative.

[a] MIC results were interpreted according to CLSI criteria for colistin % susceptible value is % intermediate; one exception is meropenem-vaborbactam for which EUCAST criteria were used. FDX MICs were read at 100% inhibition tested in CAMHB and FDC MICs were read at significant reduction tested in ID-CAMHB. As breakpoints are not available for FDX, % values represent % of MIC values for FDX ≤ 4 mg/L

[b] Xeruborbactam was tested at fixed 4 mg/L, while the concentration of cefiderocol was varied.

[c] MDR was defined as resistance to at least one drug in three or more antimicrobial categories. XDR was defined as resistance to all but ≤2 antimicrobial categories

[d] C/T-NS, isolates non-susceptible to ceftolozane-tazobactam based on CLSI criteria (MIC values ≥8 mg/L). Carbapenem-NS, isolates non-susceptible to imipenem and/or meropenem based on CLSI criteria (MIC values ≥4 mg/L).

[e] Includes bla_{GES-5} (1), bla_{GES-59} (1), bla_{IMP-7} (1), bla_{NDM-1} (24), bla_{VIM-1} (3), bla_{VIM-2} (10), bla_{VIM-4} (1), and bla_{VIM-5} (1) genes.

IDWeek 2026, October 21–24, 2026, Washington, D.C.