IDWeek 2026 | Poster #P-167

Activity of cefiderocol-xeruborbactam and comparator agents against global *Acinetobacter baumannii-calcoaceticus* species complex isolates

John H. Kimbrough[1], Krisztina M. Papp-Wallace[1], and Mariana Castanheira[1]

[1]Element Iowa City/JMI Laboratories, North Liberty, Iowa

Introduction

- *Acinetobacter baumannii-calcoaceticus* species complex (AB) infections are challenging to treat due to prolific antimicrobial resistance in this group, leaving few therapeutic options.
- Cefiderocol-xeruborbactam (FDX) is under development for AB infection treatment, including carbapenem-nonsusceptible isolates (CNS-AB).
- We examined the population structure and resistance patterns, including relevant resistance gene carriage, of AB collected in United States and European medical centers in 2024 and assessed the activity of FDX and comparator agents against these isolates.

Materials and Methods

- AB clinical isolates (*n* = 1,307) were collected from patients (1/infection episode) hospitalized in 93 medical centers across 20 countries.
- Identification was performed by matrix-assisted laser desorption ionization-time of flight mass spectrometry (Bruker Daltonics, Bremen, Germany).
- Isolates were susceptibility tested by broth microdilution using Clinical and Laboratory Standards Institute (CLSI) guidelines.
- Frozen-form broth microdilution panels, manufactured by Element Iowa City (JMI Laboratories) (North Liberty, IA, USA), contained cation-adjusted Mueller-Hinton broth (CAMHB) for cefiderocol-xeruborbactam (fixed 4 mg/L) and comparator agents; cefiderocol was tested in iron-depleted CAMHB per CLSI guidelines.
- Cefiderocol-xeruborbactam activity was interpreted at ≤4 mg/L and comparator agents were interpreted according to CLSI criteria (S, susceptible; NS, nonsusceptible); colistin utilized the intermediate (I) breakpoint (CLSI).
- CNS-AB (meropenem and/or imipenem MIC ≥4 mg/L; inclusive of CLSI intermediate and resistant isolates) were subjected to genome sequencing; screening for carbapenemase-encoding genes and multi-locus sequence typing (MLST; Pasteur scheme) were performed *in silico.*

Acknowledgments

The authors would like to thank Anthony Pannullo for assistance in generating the minimum-spanning tree and Gauri Deshpande and Jill Arends for editorial assistance. This study at Element Iowa City/JMI Laboratories was supported by Qpex Biopharma (San Diego, CA). Element Iowa City/JMI Laboratories received compensation fees for services in relation to preparing the poster, which was funded by Qpex. This project has been funded in whole or in part with federal funds from the Department of Health and Human Services; Administration for Strategic Preparedness and Response; Center for the Biomedical Advanced Research and Development Authority (BARDA), under OTA number HHSO100201600026C.

References

CLSI. 2026. M100Ed36. Performance standards for antimicrobial susceptibility testing: 36th informational supplement. Clinical and Laboratory Standards Institute.

Results

- Overall, 41.2% of AB were CNS, with higher representation among European medical centers (50.5%) compared to North American (28.8%) medical centers (Figure 1).
 - Cefiderocol-xeruborbactam demonstrated the highest activity against CNS-AB (98.5%) isolates whereas comparator activity varied widely (meropenem, 0.2%-S; colistin, 92.8%-I; Table 1).
- 93.7% of CNS-AB carried acquired carbapenemase genes and cefiderocol-xeruborbactam inhibited 98.6% of these isolates, with all other agents active against <93% of carbapenemase-producers (Figure 1, Table 1).
 - Excluding isolates with *bla*$_{NDM}$ and *bla*$_{PER}$ genes, cefiderocol-xeruborbactam was the most active agent against CNS-AB carrying *bla*$_{OXA-23}$-like (98.9%) and *bla*$_{OXA-24}$-like (98.1%) carbapenemase genes, and isolates with *bla*$_{NDM}$ were similarly well-inhibited (93.1%) (Table 1); comparator agents displayed lower activity against all molecular subsets.
 - Xeruborbactam restored the activity of cefiderocol against CNS-AB with *bla*$_{PER}$ genes (100% inhibition) compared to 4.8% activity against cefiderocol alone.
- Cefiderocol (5.7% overall) and sulbactam-durlobactam (3.9% overall) NS rates were lower than CNS rates, and geographic differences were less pronounced compared to CNS rates (cefiderocol, 4.3–6.8%; sulbactam-durlobactam, 2.1–5.2%; Figure 1); only 23 AB isolates produced cefiderocol-xeruborbactam MIC values >4 mg/L (1.8%).
 - Cefiderocol-xeruborbactam inhibited 84.0% and 90.2% of cefiderocol- and sulbactam-durlobactam-nonsusceptible isolates, respectively, at ≤4 mg/L (Table 1).
- Among CNS-AB, sequence type (ST) 2 dominated (69.7% overall; 56.5% USA/75.6% Europe; including like variants), with smaller, geographically constrained ST groups distributed across regions (Figure 2).
 - Cefiderocol-xeruborbactam was the most active agent against ST2 CNS-AB (98.7% inhibited) compared to cefiderocol (87.7%), sulbactam-durlobactam (92.8%), or colistin (91.7%) and this elevated activity was present across regions (Table 2).
- Cefiderocol-xeruborbactam, cefiderocol, and sulbactam-durlobactam inhibited identical percentages of US-restricted ST499 (97.6%) and ST1091 (100.0%) isolates.
- Cefiderocol-xeruborbactam displayed potent activity against predominant European clones, inhibiting 100.0% of isolates from ST3, ST85, ST636, and ST78 whereas the activity of cefiderocol and sulbactam-durlobactam ranged from 96.2% against ST3 to 0.0–33.3% against ST85 (Table 2).
- Only colistin (92.3%) inhibited a higher proportion of ST570 isolates than cefiderocol-xeruborbactam (84.6%); cefiderocol and sulbactam-durlobactam displayed limited activity (46.2% susceptible) against ST570 isolates (Table 2).

Figure 1. Regional representation of resistance patterns among *A. baumannii-calcoaceticus* (AB) complex isolates collected in 2024 from US (gold) and European (purple) medical centers (left) and the frequency of acquired resistance genes among carbapenem-nonsusceptible AB (CNS-AB; right). The numbers in parentheses indicate all isolates from a region and the number of CNS-AB, respectively.

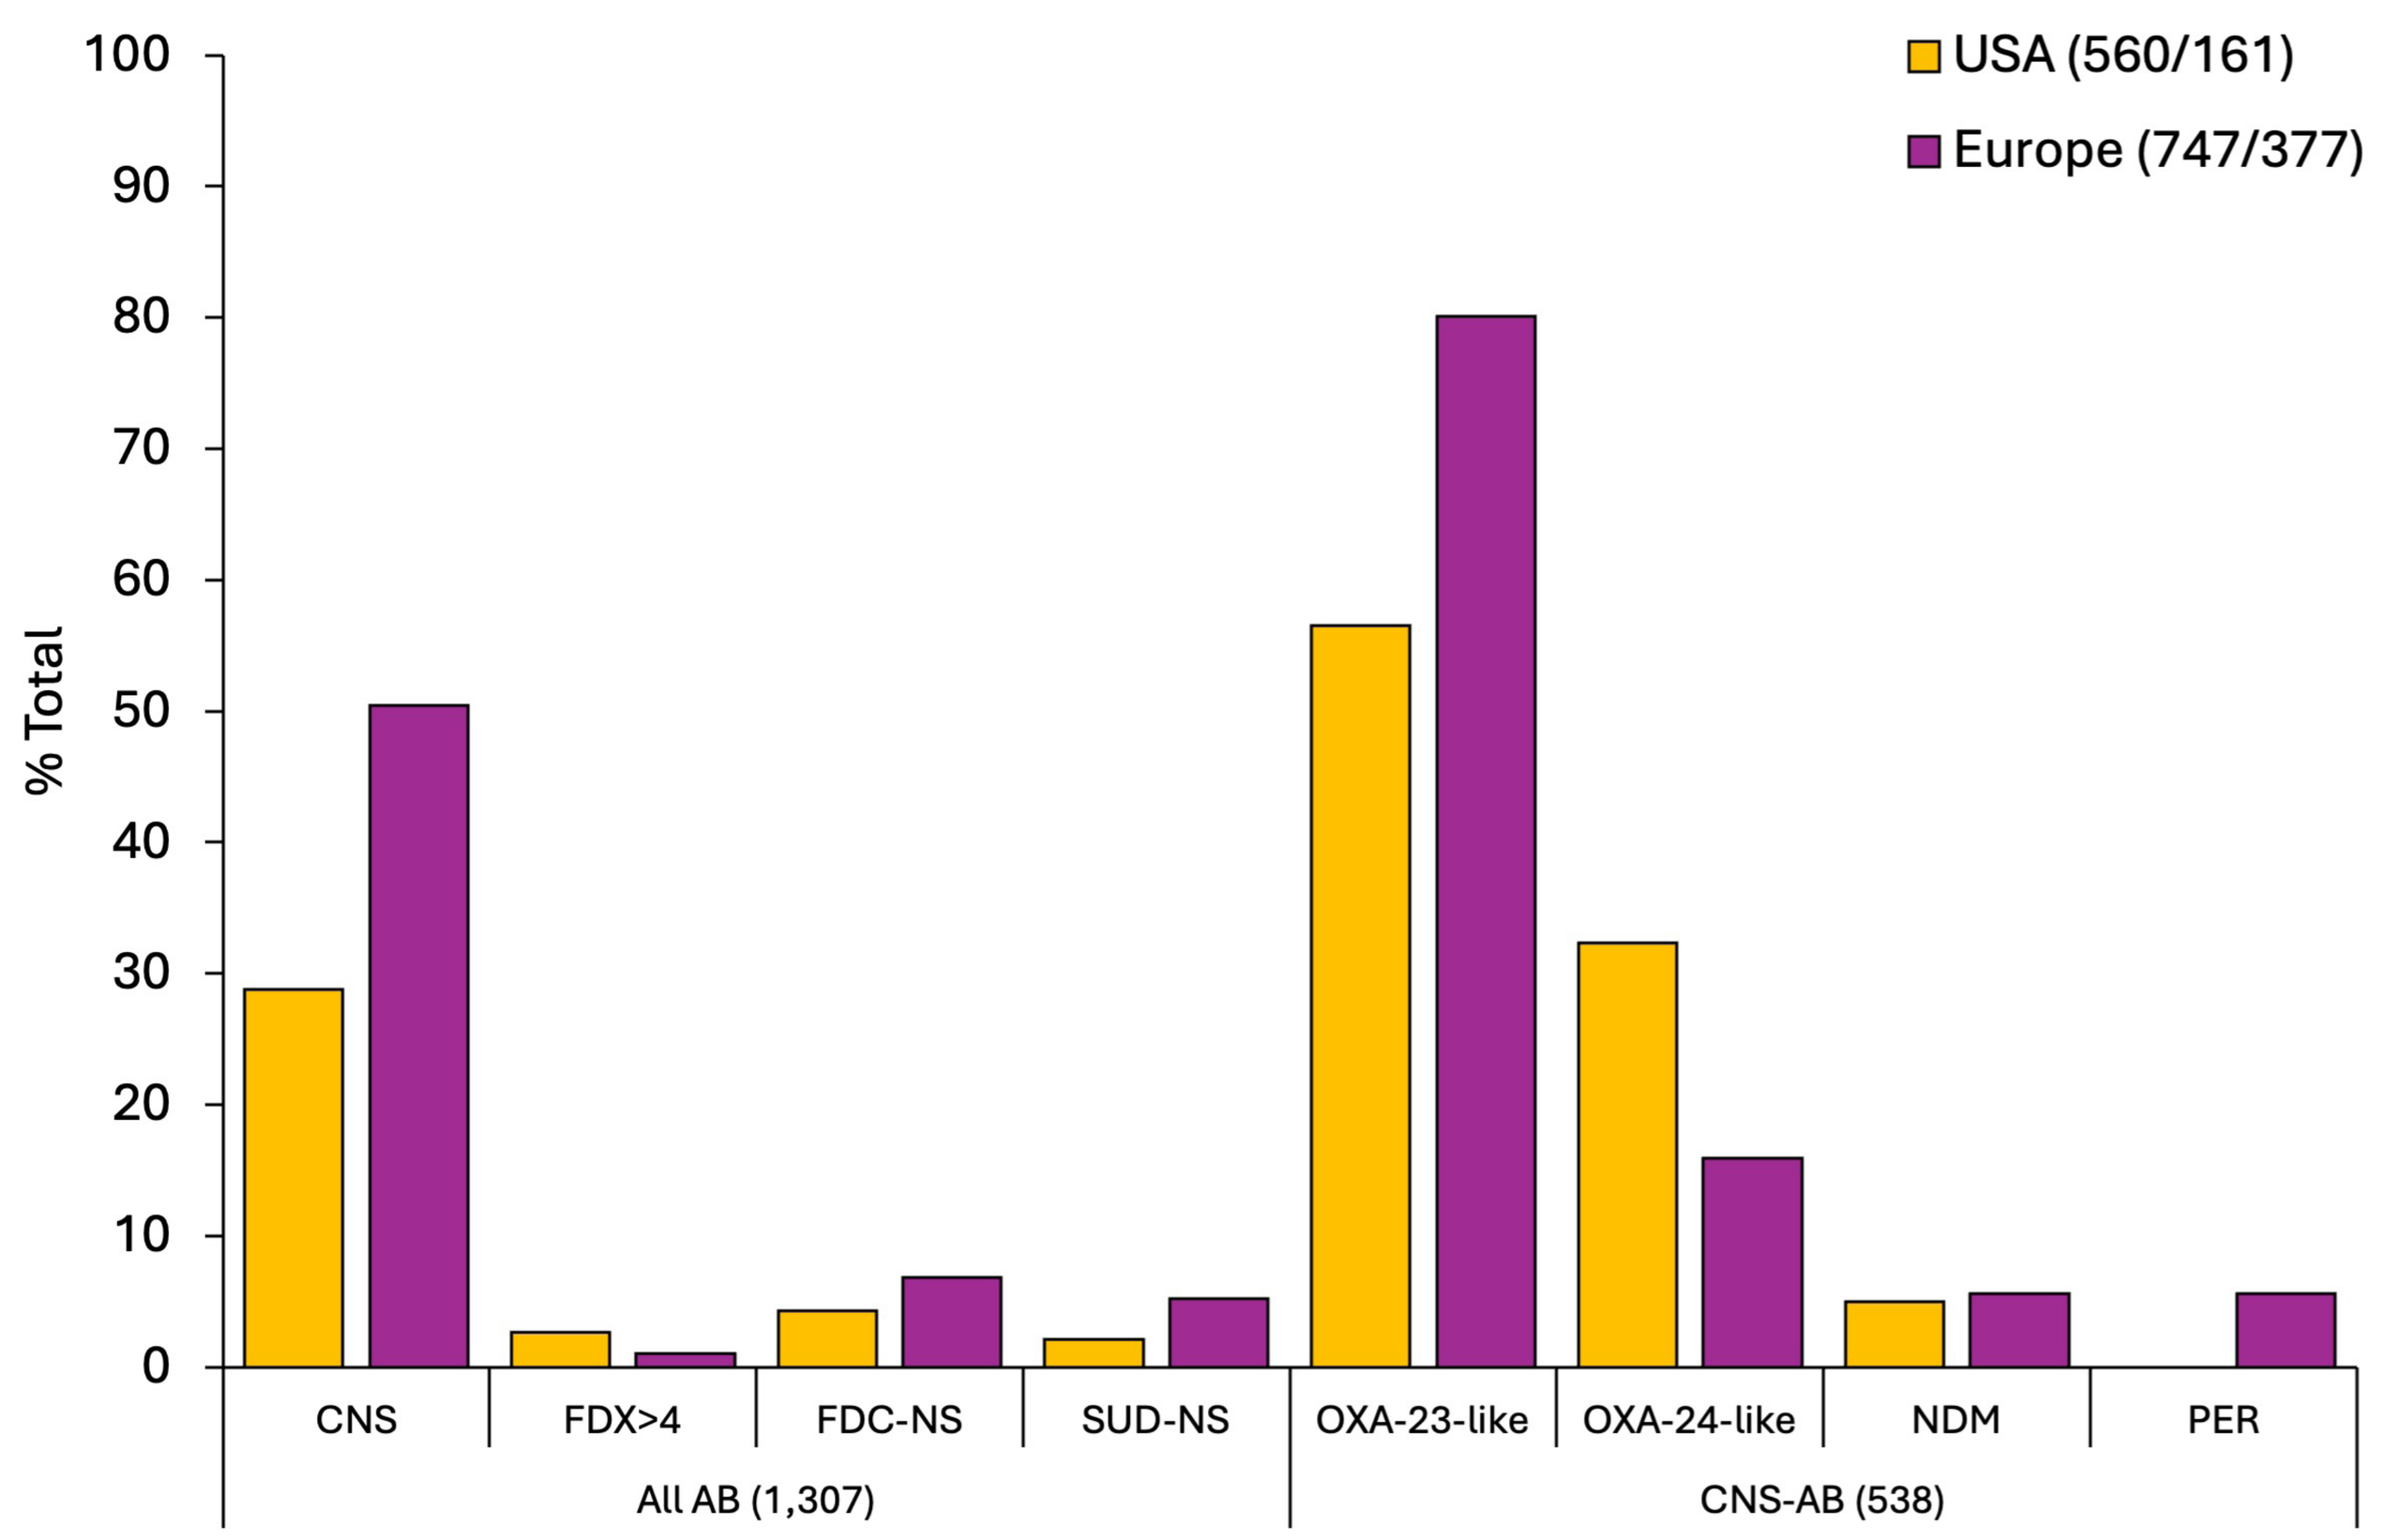
USA (560/161)
Europe (747/377)
% Total
100
90
80
70
60
50
40
30
20
10
0
CNS
FDX>4
FDC-NS
SUD-NS
OXA-23-like
OXA-24-like
NDM
PER
All AB (1,307)
CNS-AB (538)

Table 1. Activity of cefiderocol-xeruborbactam and comparator agents against *A. baumannii-calcoaceticus* complex isolates, including phenotypic and genotypic resistance subsets

Profile (*n*)	% Inhibited ($MIC_{50/90}$)[a] FDX[b]	FDC	SUD	MEM	COL[c]	A/S	TMP	LVX
All AB (1,307)	98.2 (0.25/1)	94.3 (0.12/2)	96.1 (1/2)	58.9 (1/>32)	96.3 (0.5/1)	56.5 (4/>64)	65.3 (0.5/>4)	57.0 (0.25/>8)
CNS-AB (538)	98.5 (0.5/1)	87.7 (0.25/8)	90.5 (2/4)	0.2 (>32/>32)	92.8 (0.5/2)	5.0 (64/>64)	23.4 (>4/>4)	3.0 (>8/>8)
Acquired CPase (504)	98.6 (0.5/1)	87.5 (0.25/8)	90.3 (2/4)	0.2 (>32/>32)	92.5 (0.5/2)	1.4 (64/>64)	22.0 (>4/>4)	2.0 (>8/>8)
OXA-23-like (354[d])	98.9 (0.5/1)	95.5 (0.25/4)	94.9 (2/4)	0.0 (>32/>32)	91.4 (0.5/2)	0.3 (64/>64)	24.0 (>4/>4)	1.7 (>8/>8)
OXA-24-like (106[d])	98.1 (0.5/2)	93.4 (0.25/4)	96.2 (1/4)	0.0 (>32/>32)	97.2 (0.5/1)	5.7 (32/>64)	22.6 (>4/>4)	1.9 (>8/>8)
NDM (29)	93.1 (1/4)	27.6 (8/>64)	0.0 (>32/>32)	0.0 (>32/>32)	89.7 (0.5/4)	0.0 (>64/>64)	17.2 (>4/>4)	3.4 (>8/>8)
PER (21)	100.0 (0.25/0.5)	4.8 (>64/>64)	90.5 (1/4)	0.0 (>32/>32)	100.0 (0.5/0.5)	0.0 (64/>64)	0.0 (>4/>4)	0.0 (>8/>8)
FDC-NS (75)	84.0 (1/8)	0.0 (32/>64)	65.3 (2/>32)	12.0 (>32/>32)	89.3 (0.5/4)	6.7 (64/>64)	18.7 (>4/>4)	9.3 (>8/>8)
SUD-NS (51)	90.2 (1/2)	49.0 (8/>64)	0.0 (>32/>32)	0.0 (>32/>32)	92.2 (0.5/2)	0.0 (>64/>64)	13.7 (>4/>4)	3.9 (>8/>8)

Abbreviations: MIC, minimal inhibitory concentration; FDX, cefiderocol-xeruborbactam; FDC, cefiderocol; SUD, sulbactam-durlobactam; MEM, meropenem; COL, colistin; A/S, ampicillin-sulbactam; TMP, trimethoprim-sulfamethoxazole; LVX, levofloxacin; AB, *A. baumannii-calcoaceticus* species complex; CNS, carbapenem nonsusceptible; CPase, carbapenemase; NDM, New Delhi metallo-β-lactamase; PER, *Pseudomonas* extended resistant
[a]CLSI susceptible [b]% inhibited at ≤4 mg/L [c]CLSI intermediate [d]bla$_{NDM}$/bla$_{PER}$ negative

Figure 2. Minimum-spanning tree of multi-locus sequence types (ST) observed among carbapenem-nonsusceptible *Acinetobacter baumannii-calcoaceticus* complex isolates. Bubbles represent unique STs connected by lines with thickness denoting the number of differences between linked ST groups across alleles used within the Pasteur scheme. Numbers indicate each ST (*n*≥7) and the size of each bubble corresponds to the log-transformed isolate *n*.

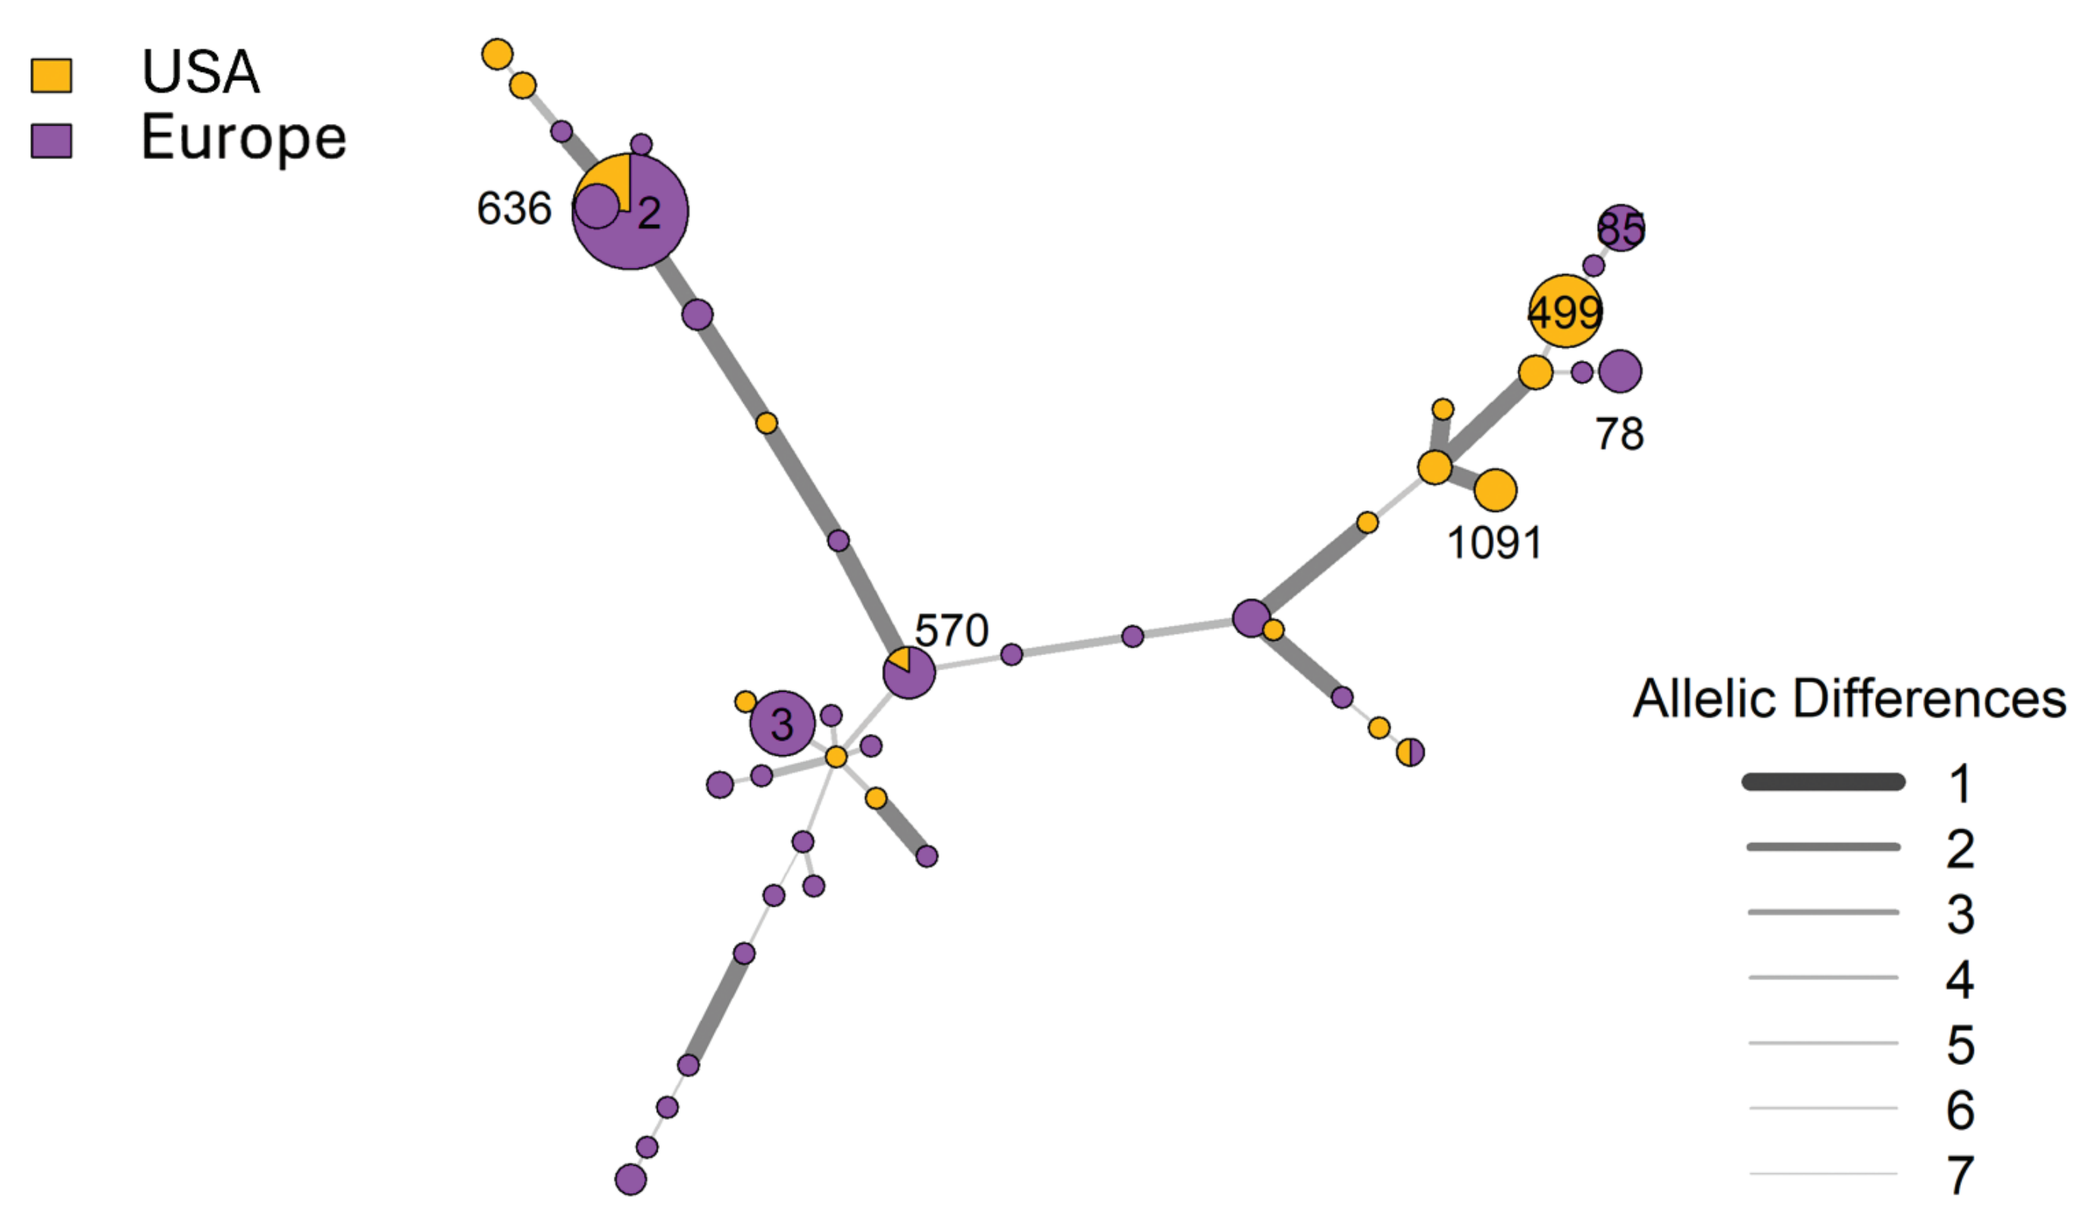
USA
Europe
636
2
85
499
78
1091
570
3
Allelic Differences
1
2
3
4
5
6
7

Table 2. Activity of cefiderocol-xeruborbactam and comparator agents against prominent CNS-*A. baumannii-calcoaceticus* complex STs

Profile (*n*)[b]	% Inhibited ($MIC_{50/90}$)[a] FDX[c]	FDC	SUD	MEM	COL[d]	A/S	TMP	LVX
CNS-AB (538)	98.5 (0.5/1)	87.7 (0.25/8)	90.5 (2/4)	0.2 (>32/>32)	92.8 (0.5/2)	5.0 (64/>64)	23.4 (>4/>4)	3.0 (>8/>8)
ST2 (375)	98.7 (0.5/1)	87.7 (0.25/8)	92.8 (2/4)	0.0 (>32/>32)	91.7 (0.5/2)	3.7 (64/>64)	21.1 (>4/>4)	0.3 (>8/>8)
USA (90)	98.2 (0.25/2)	85.6 (0.5/16)	91.1 (2/4)	0.0 (>32/>32)	94.4 (0.5/1)	10.0 (64/>64)	40.0 (>4/>4)	1.1 (>8/>8)
Europe (285)	98.6 (0.5/1)	88.4 (0.25/8)	93.3 (2/4)	0.0 (>32/>32)	90.9 (0.5/2)	1.8 (64/>64)	15.1 (>4/>4)	0.0 (>8/>8)
ST499 (41)	97.6 (0.5/2)	97.6 (0.12/2)	97.6 (1/4)	0.0 (>32/>32)	95.1 (0.5/1)	2.4 (32/64)	43.9 (>4/>4)	2.4 (>8/>8)
ST3 (26)	100.0 (0.5/1)	96.2 (0.12/4)	96.2 (1/4)	0.0 (32/>32)	100.0 (0.5/1)	0.0 (64/>64)	19.1 (>4/>4)	0.0 (>8/>8)
ST570 (13)	84.6 (1/8)	46.2 (8/>64)	46.2 (>32/>32)	0.0 (32/>32)	92.3 (0.5/0.5)	0.0 (>64/>64)	7.7 (>4/>4)	0.0 (>8/>8)
ST85 (9)	100.0 (1/—)	33.3 (8/—)	0.0 (>32/—)	0.0 (>32/—)	88.9 (0.5/—)	0.0 (>64/—)	11.1 (>4/—)	0.0 (>8/—)
ST636 (8)	100.0 (1/—)	100.0 (1/—)	100.0 (1/—)	0.0 (>32/—)	100.0 (0.5/—)	0.0 (>64/—)	0.0 (>4/—)	0.0 (8/—)
ST78 (7)	100.0 (0.25/—)	85.7 (0.5/—)	85.7 (1/—)	0.0 (>32/—)	100.0 (0.25/—)	14.3 (64/—)	0.0 (>4/—)	0.0 (>8/—)
ST1091 (7)	100.0 (0.12/—)	100.0 (2/—)	100.0 (1/—)	0.0 (>32/—)	80.0 (0.5/—)	20.0 (32/—)	0.0 (>4/—)	0.0 (>8/—)
Other ST (52)[e]	100.0 (0.25/1)	91.5 (0.12/4)	89.4 (1/8)	2.1 (32/>32)	93.6 (0.5/2)	19.1 (32/>64)	42.6 (>4/>4)	29.8 (8/>8)

Abbreviations: ST, sequence type; MIC, minimal inhibitory concentration; FDX, cefiderocol-xeruborbactam; FDC, cefiderocol; SUD, sulbactam-durlobactam; MEM, meropenem; COL, colistin; A/S, ampicillin-sulbactam; TMP, trimethoprim-sulfamethoxazole; LVX, levofloxacin; AB, *A. baumannii-calcoaceticus* species complex; CNS, carbapenem nonsusceptible
[a]CLSI susceptible; MIC_{90} values were only calculated when n≥10 [b]Only STs >1% [c]% inhibited at ≤4 mg/L [d]CLSI intermediate [e]Includes 5 isolates from ST1 and ST406/406-like each, ST2512 (n=4), 3 isolates from ST15, ST492, and ST788-like, 2 isolates each from ST158, ST164, ST1022/1022-like, and ST1143, and singletons of ST25, ST32, ST79, ST108, ST193, ST207, ST220, ST374, ST452, ST498, ST592, ST620, ST717, ST734, ST776, ST1142, ST2494, ST629-like, ST697-like, and ST972-like; 1 isolate could not be assigned an ST group.

Conclusions

- Cefiderocol-xeruborbactam consistently demonstrated high activity across phenotypic and genotypic resistance subsets of a large collection of contemporary AB clinical isolates including those isolates carrying class D OXA-type and NDM family carbapenemase-encoding genes.
- Cefiderocol-xeruborbactam MIC values >4 mg/L were rare among AB (1.8%) and were lower than nonsusceptibility rates for all other tested agents.
- The addition of xeruborbactam largely restored the activity of cefiderocol against cefiderocol-nonsusceptible isolates, including those isolates carrying *bla*$_{PER}$ genes.
- Cefiderocol-xeruborbactam was the most active agent against the majority of predominant clones present worldwide (ST2, ST570) and those limited to the USA (ST499, ST1091) and Europe (ST3, ST85, ST78).
- The results of this study support the continued development of cefiderocol-xeruborbactam for the treatment of infections caused by AB from diverse clonal backgrounds with complex resistance mechanisms nonsusceptible to many currently available treatment options.

Contact

John H. Kimbrough, Ph.D.
Element Iowa City (JMI Laboratories)
345 Beaver Kreek Centre, Suite A
North Liberty, Iowa 52317

Phone: (319) 665-3370
Fax: (319) 665-3371
Email: hank.kimbrough@element.com

Scan Me

To obtain a PDF of this poster:

Scan the QR code or visit https://www.jmilabs.com/data/posters/IDWeek2026_26-QPX-02_P3_CFDC-Xeru_ACB.pdf

Charges may apply. No personal information is stored.

IDWeek 2026, October 21–24, 2026, Washington, D.C.