Comparison of Ceftazidime-Avibactam, Ceftolozane-Tazobactam, Piperacillin-Tazobactam, and Meropenem Activities when Tested against Gram-Negative Organisms Isolated from Complicated Urinary Tract Infections

IDWeek 2019
Poster #1436

Helio S. Sader, Robert K. Flamm, Mariana Castanheira, Rodrigo E. Mendes

JMI Laboratories, North Liberty, Iowa, USA

Helio S. Sader, MD, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, IA 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio-sader@jmilabs.com

INTRODUCTION

- Ceftazidime-avibactam is approved by the United States Food and Drug Administration (US FDA) to treat hospital-acquired bacterial pneumonia, including ventilator-associated bacterial pneumonia, complicated intra-abdominal infections in combination with metronidazole, and complicated urinary tract infections (cUTIs), including pyelonephritis
- We evaluated the antimicrobial susceptibility of gram-negative bacilli causing complicated urinary tract infection (cUTI) in US medical centers, focusing on the β-lactamase inhibitor combinations ceftazidime-avibactam, ceftolozane-tazobactam, and piperacillin-tazobactam and the carbapenem meropenem

MATERIALS AND METHODS

Bacterial isolates

- A total of 4,371 gram-negative bacilli, including 4,120 *Enterobacterales*, 230 *P. aeruginosa*, and 21 *Acinetobacter* spp. isolates, were consecutively collected (1/patient) from patients with cUTI in 65 US medical centers in 2018
- Carbapenem-resistant *Enterobacterales* (CRE) isolates were defined as displaying imipenem, meropenem, and/or doripenem MIC values at ≥4 mg/L (Clinical and Laboratory Standards Institute [CLSI], 2019)
 - Imipenem was not applied to *Proteus mirabilis* or indole-positive Proteeae due to the intrinsically elevated MIC values

Susceptibility testing

- All isolates were tested for susceptibility by reference broth microdilution test method according to CLSI as part of the INFORM program
- Avibactam was provided by Allergan (Irvine, California, USA) and combined with ceftazidime (avibactam at fixed concentration of 4 mg/L) for susceptibility testing
- Ceftolozane stock solution was obtained from ThermoFisher Scientific (Cleveland, Ohio, USA) and combined with tazobactam (acquired from United States Pharmacopeia [USP]) at fixed concentration of 4 mg/L for susceptibility testing
- All other compounds were obtained from USP or Sigma-Aldrich (St. Louis, Missouri, USA)

Screening for β-lactamase-encoding genes

- *Enterobacterales* isolates displaying MIC values ≥2 mg/L for at least 2 β-lactams (ie, ceftazidime, ceftriaxone, aztreonam, or cefepime) and all CRE isolates were tested for β-lactamase-encoding genes using next-generation sequencing

RESULTS

- The most common gram-negative organisms were *Escherichia coli* (44.5%), *Klebsiella pneumoniae* (19.6%), *P. mirabilis* (6.7%), and *P. aeruginosa* (5.3%; data not shown)
- The most active agents against *Enterobacterales* were ceftazidime-avibactam (99.9% susceptible [S]), amikacin (99.7%S), and meropenem (99.4%S), whereas ceftolozane-tazobactam and piperacillin-tazobactam were active against 96.7% and 95.0% of isolates at the respective susceptible breakpoint (Table 1)
- Extended-spectrum β-lactamase (ESBL) genes were identified in 315 *Enterobacterales* (7.6%; excluding carbapenemase co-producers), including CTX-M-15 (63% of ESBL producers), other CTX-M types (25%), OXA-1/OXA-30 (41%), and SHV-type ESBLs (8%); approximately 40% of ESBL producers had ≥2 ESBL genes, mainly a CTX-M-type and an OXA-type (37% of isolates; data not shown)
- The most active agents against ESBL producers were ceftazidime-avibactam (100.0%S), amikacin (99.7%S), and meropenem (99.4%S), while ceftolozane-tazobactam and piperacillin-tazobactam were active against 90.6% and 84.8% of ESBL producers, respectively (Figure 1)
- Only ceftazidime-avibactam (87.0%S; Figure 1), colistin (87.0%S per EUCAST), and tigecycline (95.7%S) exhibited good activity against CRE
- Only 3 *Enterobacterales* isolates (0.07%) were ceftazidime-avibactam resistant and all had a metallo-β-lactamase gene (2 VIM-1 and 1 NDM-1; data not shown)
- Ceftazidime-avibactam (97.0%S; Table 1 and Figure 2) and ceftolozane-tazobactam (99.1%S) were the most active β-lactams tested against *P. aeruginosa*; other compounds with >90%S for *P. aeruginosa* were colistin (99.6%), amikacin (97.8%), tobramycin (93.5%), and ceftazidime (90.4%; Table 1)
- *Acinetobacter* spp. represented only 0.5% of the isolates collected from patients with cUTI, and the most active agents tested against these organisms were colistin (100.0%S), followed by the carbapenems imipenem and meropenem and the aminoglycosides amikacin and tobramycin, all 4 compounds with 95.2%S (data not shown)

CONCLUSIONS

- Ceftazidime-avibactam demonstrated potent activity against contemporary *Enterobacterales* isolates (n=4,120) from patients with cUTI in US hospitals, including ESBL producers and CRE
- Ceftolozane-tazobactam, piperacillin-tazobactam, and meropenem exhibited good activity against the overall *Enterobacterales* (≥95%S) collection, but ceftolozane-tazobactam and piperacillin-tazobactam displayed limited activity against ESBL producers and all 3 compounds were inactive against CRE
- Ceftazidime-avibactam and ceftolozane-tazobactam showed similar coverage (%S) against *P. aeruginosa* (97.0%S vs. 99.1%S, respectively)
- Ceftazidime-avibactam exhibited the best overall coverage (*Enterobacterales* + *P. aeruginosa*) among antimicrobial agents tested against gram-negative bacilli from patients with cUTI in US hospitals during 2018

Figure 1 Antimicrobial susceptibility of *Enterobacterales* isolated from patients with complicated urinary tract infections in US medical centers (INFORM program, 2018)

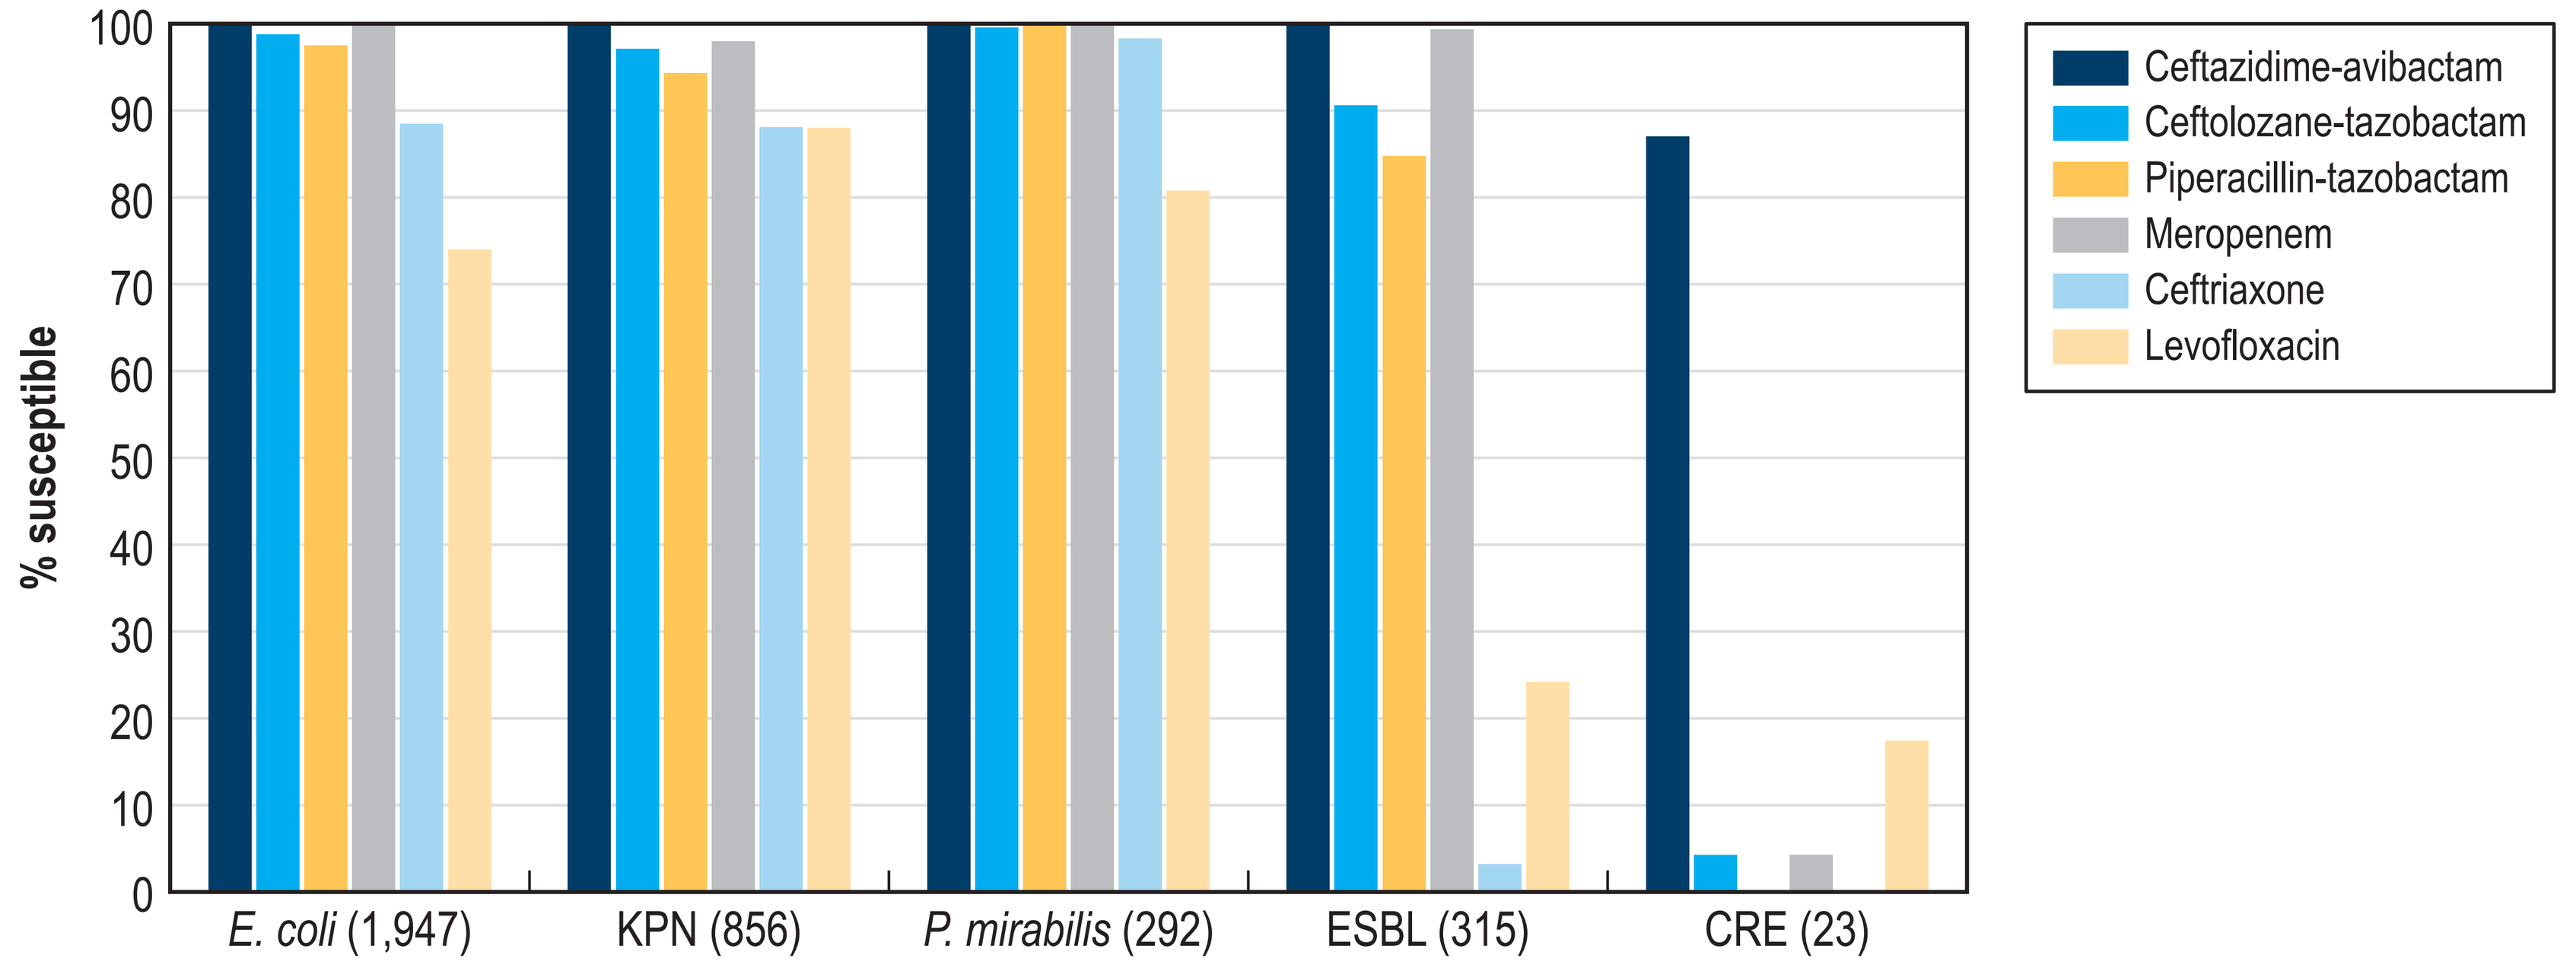
% susceptible
0
10
20
30
40
50
60
70
80
90
100
E. coli (1,947)
KPN (856)
P. mirabilis (292)
ESBL (315)
CRE (23)
Ceftazidime-avibactam
Ceftolozane-tazobactam
Piperacillin-tazobactam
Meropenem
Ceftriaxone
Levofloxacin

KPN, *K. pneumoniae*; ESBL, extended-spectrum β-lactamases (excluding carbapenemase producing strains); CRE, carbapenem-resistant *Enterobacterales*.

Table 1 Antimicrobial activity of ceftazidime-avibactam and comparator agents tested against *Enterobacterales* and *P. aeruginosa* isolated from patients with complicated urinary tract infections in US medical centers (2018)

Organism/organism group (no. of isolates) Antimicrobial agent	MIC_{50} (mg/L)	MIC_{90} (mg/L)	CLSI[a]	
			%S	%R
Enterobacterales (4,120)				
Ceftazidime-avibactam	0.12	0.25	99.9	0.1
Ceftolozane-tazobactam	0.25	1	96.7	2.4
Piperacillin-tazobactam	2	8	95.0	2.3
Ceftriaxone	≤0.06	>8	86.8	12.6
Meropenem	0.03	0.06	99.4	0.4
Levofloxacin	0.06	8	80.9	16.7
Amikacin	2	4	99.7	0.2
Colistin	0.12	>8	85.2[b]	14.8[b]
Pseudomonas aeruginosa (230)				
Ceftazidime-avibactam	2	4	97.0	3.0
Ceftolozane-tazobactam	1	2	99.1	0.9
Piperacillin-tazobactam	4	32	85.2	5.7
Ceftazidime	2	8	90.4	6.1
Meropenem	0.5	4	86.1	9.1
Levofloxacin	0.5	32	65.5	25.3
Tobramycin	0.5	2	93.5	6.5
Colistin	0.5	1	99.6	0.4

[a] Criteria as published by CLSI 2019.
[b] Per EUCAST 2019 criteria.

Figure 2 Ceftazidime-avibactam susceptibility of *P. aeruginosa* isolated from patients with complicated urinary tract infections in US medical centers (INFORM program, 2018)

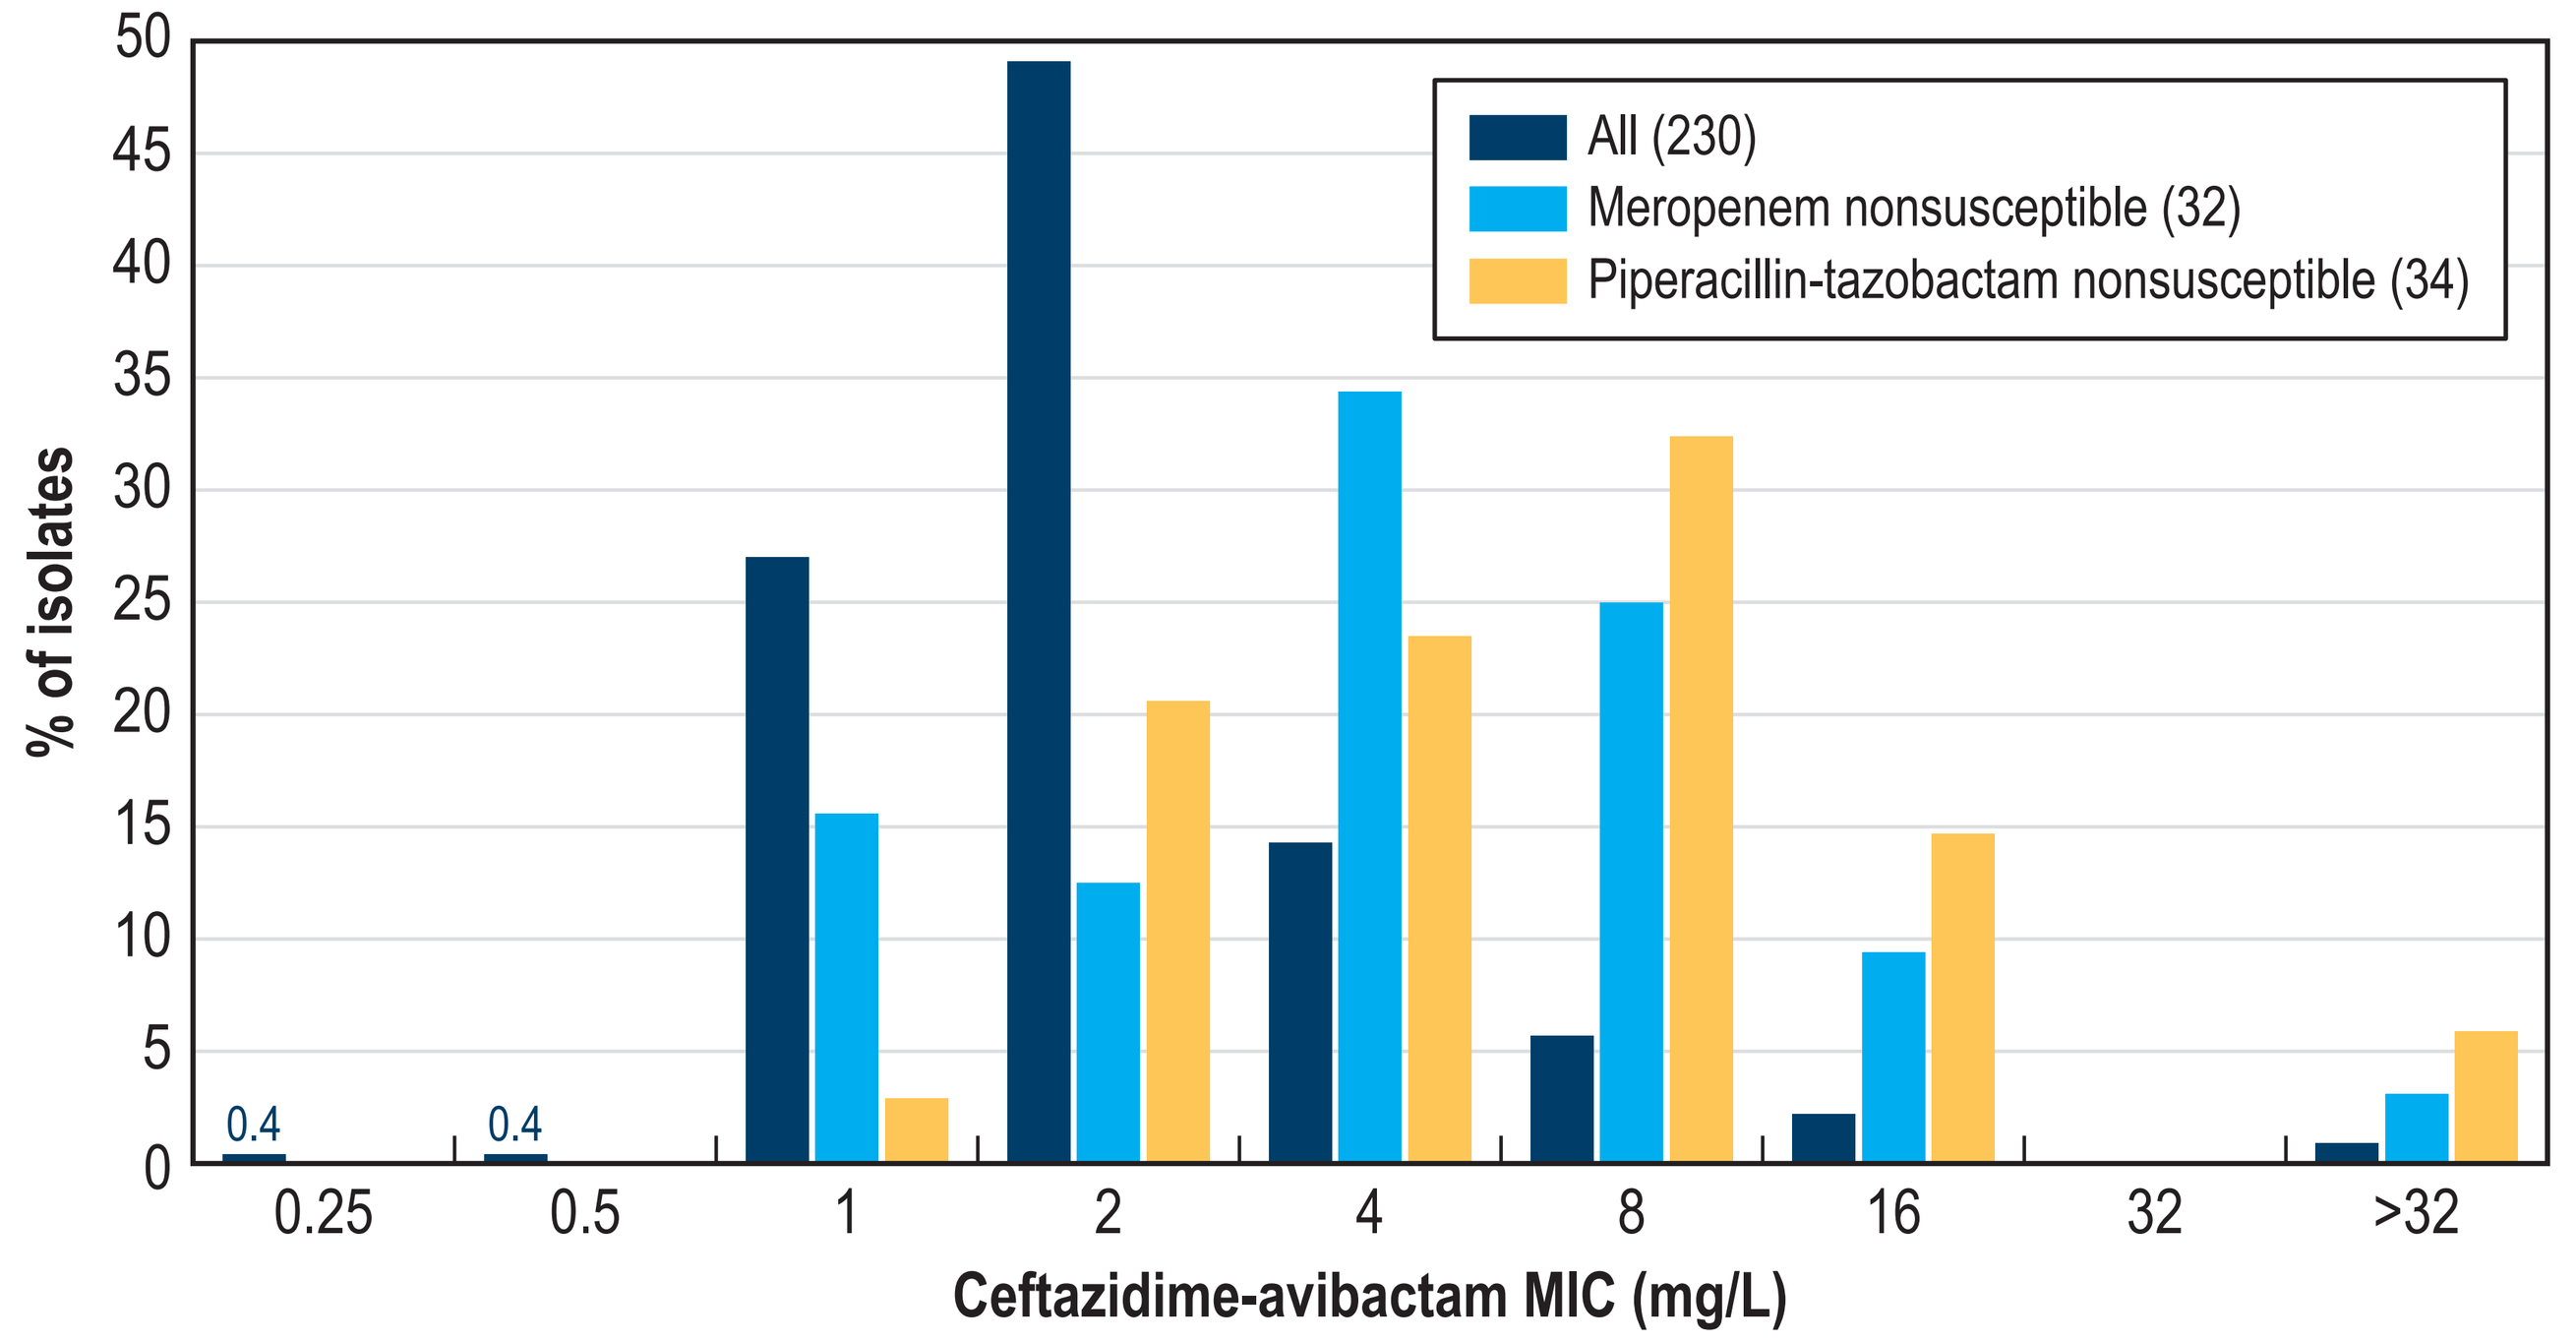
% of isolates
0
5
10
15
20
25
30
35
40
45
50
0.4
0.4
0.25
0.5
1
2
4
8
16
32
>32
Ceftazidime-avibactam MIC (mg/L)
All (230)
Meropenem nonsusceptible (32)
Piperacillin-tazobactam nonsusceptible (34)

ACKNOWLEDGEMENTS

The authors would like to thank all participants of the International Network for Optimal Resistance Monitoring (INFORM) program for providing bacterial isolates.

This study was supported by Allergan. Allergan was involved in the design and decision to present these results, and JMI Laboratories received compensation for services related to preparing the poster. Allergan had no involvement in the collection, analysis, or interpretation of data.

REFERENCES

1. AVYCAZ® (2016). AVYCAZ® (ceftazidime-avibactam). Allergan USA, Inc. Available at https://www.allergan.com /assets/pdf/avycaz_pi. Accessed January 2017.
2. Castanheira M, Mendes RE, Jones RN, Sader HS (2016). Changes in the frequencies of beta-lactamase genes among *Enterobacteriaceae* isolates in U.S. Hospitals, 2012 to 2014: Activity of ceftazidime-avibactam tested against beta-Lactamase-producing isolates. *Antimicrob Agents Chemother* 60: 4770-4777.
3. Clinical and Laboratory Standards Institute (2019). *M100Ed29E. Performance standards for antimicrobial susceptibility testing: 29th informational supplement.* Wayne, PA: CLSI.
4. Clinical and Laboratory Standards Institute (2018). *M07Ed11E. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard: eleventh edition.* Wayne, PA: CLSI.
5. Sader HS, Castanheira M, Duncan LR, Flamm RK (2018). Antimicrobial susceptibility of *Enterobacteriaceae* and *Pseudomonas aeruginosa* Isolates from United States medical centers stratified by infection type: Results from the International Network for Optimal Resistance Monitoring (INFORM) surveillance program, 2015-2016. *Diagn Microbiol Infect Dis* 92: 69-74.

Visit www.allergancongressposters .com/210152

Allergan

IDWeek 2019, October 2–6, 2019, Washington, DC