Poster: Saturday-335

Activity of Telavancin Against a Global Collection of *Staphylococcus aureus* Causing Bacteremia (2011-2014)

RE Mendes[1], HS Sader[1], DJ Farrell[1], JI Smart[2], RK Flamm[1], RN Jones[1]

[1] JMI Laboratories, North Liberty, Iowa, USA; [2] Theravance Biopharma US, Inc., South San Francisco, California, USA

Contact Information:
Rodrigo E. Mendes, PhD
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, Iowa 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: rodrigo-mendes@jmilabs.com

ABSTRACT

Background: Telavancin versus standard intravenous therapy is under investigation for the treatment of subjects with *S. aureus* bacteremia, including endocarditis in a Phase 3 trial. Telavancin activity was assessed against a global collection of *S. aureus* bacteremia isolates, including those responsible for endocarditis.

Methods: 4,191 *S. aureus* bacteremia isolates from a global network of hospitals were included. Isolates were submitted to a central laboratory as part of a surveillance program (2011-2014). Identification was confirmed and susceptibility testing performed by CLSI methods. MIC interpretation of telavancin results used the USA FDA, CLSI and EUCAST approved criteria.

Results: Overall, *S. aureus* isolates (100.0% telavancin susceptible) had telavancin MIC_{50}, MIC_{90} and MIC_{100} results of 0.03, 0.06 and 0.12 µg/ml, respectively. Equivalent MICs ($MIC_{50/90}$, 0.03/0.06 µg/ml) were obtained for methicillin-susceptible (MSSA) and -resistant (MRSA) isolates, as well as MRSA from community and nosocomial origins. Telavancin ($MIC_{50/90}$, 0.03/0.06 µg/ml) had similar potency against MRSA from North America and Europe, while isolates from the Asia-Pacific (APAC) and Latin America regions had slightly higher MIC_{50} values ($MIC_{50/90}$, 0.06/0.06 µg/ml). MRSA with vancomycin MICs of 2-4 µg/ml had telavancin MICs ($MIC_{50/90}$, 0.06/0.12 µg/ml) 2-fold higher than those with vancomycin MICs at ≤1 µg/ml ($MIC_{50/90}$, 0.03/0.06 µg/ml), but telavancin still inhibited all isolates at the susceptible breakpoint of ≤0.12 µg/ml. *S. aureus* causing endocarditis were inhibited by telavancin ($MIC_{50/90}$, 0.03/0.06 µg/ml) at ≤0.12 µg/ml (100.0% susceptible). Overall, telavancin was 8-fold more potent than daptomycin ($MIC_{50/90}$, 0.25/0.5 µg/ml) and 16- to 32-fold more potent than linezolid ($MIC_{50/90}$, 1/1 µg/ml) and vancomycin ($MIC_{50/90}$, 1/1 µg/ml) against MRSA.

Conclusions: Telavancin (100.0% susceptible) demonstrated potent activity against this global and contemporary collection of *S. aureus* causing bacteremia, including endocarditis. These *in vitro* results support the investigation of telavancin for the treatment of *S. aureus* bacteremia.

INTRODUCTION

Staphylococcus aureus is the second most common cause of bloodstream infection (BSI), and is the most important cause of BSI-associated death. Population-based studies conducted in many regions around the world have identified incidence rates of 15–40 per 100,000 population per year, with case-fatality rates of approximately 15–25%. In 2015, *S. aureus* represented 44.3% (45.4% MRSA) of all Gram-positive isolates causing bacteremia in USA hospitals during the SENTRY Antimicrobial Surveillance Program. Although *S. aureus* represent an important cause of BSI, the treatment of invasive MRSA infections has relied significantly on vancomycin. However, the reports of poor clinical outcomes of BSI caused by MRSA isolates displaying elevated vancomycin MIC results (i.e. 2 mg/L) has prompted the call for alternative agents.

Telavancin is a lipoglycopeptide antimicrobial agent with a dual mechanism of action that involves both inhibition of peptidoglycan synthesis and disruption of bacterial cell membrane function, which provides telavancin with potent activity against a broad spectrum of Gram-positive isolates. Telavancin is approved for the treatment of complicated skin and skin structure infections, and hospital-acquired and ventilator-associated bacterial pneumonia. Additional clinical trials will evaluate telavancin in expanded therapeutic uses, such as for the treatment of *S. aureus* bacteremia, including endocarditis, and osteomyelitis. This *in vitro* study evaluates telavancin activity against a global collection of *S. aureus* bacteremia isolates, including those responsible for endocarditis.

MATERIALS AND METHODS

Bacterial strain collection. A total of 4,191 *S. aureus* (1,490 MRSA) bacteremia clinical isolates from a global network of hospitals in North America (2,150), Europe (1,283), Latin America (473) and Asia-Pacific (APAC; 285) regions were included. All isolates were deemed responsible for bacteremia, including BSI and/or endocarditis. Isolates were submitted to a central monitoring laboratory (JMI Laboratories, North Liberty, Iowa USA), as part of the SENTRY Antimicrobial Surveillance Program. Isolates were initially identified by the participating laboratory and identification confirmed by the reference monitoring laboratory by standard algorithms and supported by MALDI-TOF-MS (Bruker Daltonics, Bremen, Germany).

Antimicrobial susceptibility test methods. Isolates were tested for susceptibility by broth microdilution following the Clinical and Laboratory Standards Institute (CLSI) M07-A10 document. Testing was performed using panels manufactured by Thermo Fisher Scientific (Oakwood Village, Ohio, USA). These panels provide telavancin results equivalent to the CLSI-approved broth microdilution method supplemented with 0.002% polysorbate-80. Bacterial inoculum density was monitored by colony counts to assure an adequate number of cells for each testing event. Affirmation of the MIC values was performed by concurrent testing of CLSI-recommended quality control reference strains (*S. aureus* ATCC 29213 and *Enterococcus faecalis* ATCC 29212). MIC breakpoint interpretation used current USA FDA, CLSI and EUCAST approved criteria. MRSA isolates displaying resistance phenotype to at least three classes of drugs other than β-lactam agents were considered multidrug-resistant (MDR). The CDC criteria was utilized for definition of healthcare-associated (HA) and community-acquired (CA) isolates.

RESULTS

- Overall, *S. aureus* isolates (100.0% telavancin-susceptible) had telavancin MIC_{50}, MIC_{90} and MIC_{100} results of 0.03, 0.06 and 0.12 µg/ml, respectively. Equivalent MICs ($MIC_{50/90}$, 0.03/0.06 µg/ml) were obtained for methicillin-susceptible (MSSA) and -resistant (MRSA) isolates as well as MRSA from CA and HA origins (**Table 1**).
- Telavancin ($MIC_{50/90}$, 0.03/0.06 µg/ml) had similar potency against all *S. aureus* or the MRSA subsets from North America and Europe, while isolates from the Asia-Pacific (APAC) and Latin America regions had slightly higher MIC_{50} values ($MIC_{50/90}$, 0.06/0.06 µg/ml; **Table 1**).
- MRSA with vancomycin MIC values of 2-4 µg/ml and the MDR subset had telavancin MIC_{50} results of 0.06 µg/ml which was 2-fold higher than the telavancin MIC_{50} results (MIC_{50}, 0.03 µg/ml) for the isolates with vancomycin MIC values at ≤1 µg/ml or a non-MDR phenotype. Even though the MIC_{50} was higher in this resistant subgroup, telavancin still inhibited all isolates at the susceptible breakpoint of ≤0.12 µg/ml (**Table 1**).
- Daptomycin ($MIC_{50/90}$, 0.5/1 µg/ml; **Table 2**) also demonstrated higher MIC results when tested against MRSA exhibiting vancomycin MIC values of 2-4 µg/ml compared with those isolates with vancomycin MIC values at ≤1 µg/ml (data not shown).
- *S. aureus* causing endocarditis were inhibited by telavancin ($MIC_{50/90}$, 0.03/0.06 µg/ml) at ≤0.12 µg/ml (100.0% susceptible; **Table 1**). Overall, telavancin showed MIC_{50} results 8-fold lower than daptomycin ($MIC_{50/90}$, 0.25/0.5 µg/ml) and up to 32-fold lower than vancomycin ($MIC_{50/90}$, 1/1 µg/ml) against MRSA (**Figure 1 and Table 2**).
- Similarly, telavancin MIC results ($MIC_{50/90}$, 0.06/0.06 µg/ml) were 4- to 8-fold lower than those obtained by daptomycin ($MIC_{50/90}$, 0.25/0.5 µg/ml) and 16-fold lower than vancomycin ($MIC_{50/90}$, 1/1 µg/ml) against the MRSA MDR subset (**Table 2**).
- When tested against the MRSA subset displaying decreased susceptibility to vancomycin (MIC, 2-4 µg/ml), telavancin ($MIC_{50/90}$, 0.06/0.12 µg/ml) and daptomycin ($MIC_{50/90}$, 0.5/1 µg/ml) were the most potent agents; however, telavancin was 8-fold more potent than daptomycin (**Table 2**).

Table 1. Antimicrobial activity and MIC distributions for telavancin when tested against *S. aureus* clinical isolates, as part of the international telavancin surveillance program.

Parameter[a] (number tested)	MIC (µg/ml) 50%	MIC (µg/ml) 90%	Number (cumulative %) inhibited at telavancin MIC (µg/ml) of: ≤0.015	0.03	0.06	0.12
Infection type						
BSI (4149)	0.03	0.06	166 (4.0)	2444 (62.9)	1520 (99.5)	19 (100.0)
Endocarditis (42)	0.03	0.06	3 (7.1)	21 (57.1)	18 (100.0)	
Origin						
CA-MRSA (828)	0.03	0.06	24 (2.9)	482 (61.1)	312 (98.8)	10 (100.0)
HA-MRSA (552)	0.03	0.06	15 (2.7)	284 (54.2)	250 (99.5)	3 (100.0)
Phenotype						
MSSA (2,701)	0.03	0.06	127 (4.7)	1632 (65.1)	938 (99.9)	4 (100.0)
MRSA (1,490)	0.03	0.06	42 (2.8)	833 (58.7)	600 (99.0)	15 (100.0)
MDR (569)	0.06	0.06	8 (1.4)	264 (47.8)	285 (97.9)	12 (100.0)
Non-MDR (921)	0.03	0.06	34 (3.7)	569 (65.5)	315 (99.7)	3 (100.0)
Vancomycin MIC ≤1 µg/ml (4,140)	0.03	0.06	29 (3.2)	582 (67.5)	289 (99.4)	5 (100.0)
Vancomycin MIC = 2-4 µg/ml (51)	0.06	0.12	1 (3.0)	4 (15.2)	22 (81.8)	6 (100.0)
Region						
North America (2,150)	0.03	0.06	102 (4.7)	1374 (68.7)	662 (99.4)	12 (100.0)
MRSA (938)	0.03	0.06	30 (3.2)	586 (65.7)	311 (98.8)	11 (100.0)
Europe (1,283)	0.03	0.06	52 (4.1)	798 (66.3)	430 (99.8)	3 (100.0)
MRSA (290)	0.03	0.06	10 (3.4)	174 (63.4)	105 (99.7)	1 (100.0)
Latin America (473)	0.06	0.06	13 (2.7)	206 (46.3)	250 (99.2)	4 (100.0)
MRSA (175)	0.06	0.06	1 (0.6)	47 (27.4)	124 (98.3)	3 (100.0)
APAC (285)	0.06	0.06	2 (0.7)	87 (31.2)	196 (100.0)	
MRSA (87)	0.06	0.06	1 (1.1)	26 (31.0)	60 (100.0)	

a. BSI=bloodstream infection; MSSA = methicillin-susceptible *S. aureus*; MRSA = methicillin-resistant *S. aureus*; CA-MRSA=community-acquired MRSA; HA-MRSA; healthcare-associated MRSA; Origin of isolate was defined based on CDC criteria; MDR = multidrug-resistant, defined as MRSA (methicillin [oxacillin]-resistant) resistant to three or more drug classes in addition to β-lactam agents.

Table 2. Antimicrobial activity of telavancin and comparator agents tested against a global collection of *S. aureus* clinical isolates responsible for bacteremia.

Organism (number tested)/ Antimicrobial agent[a]	MIC (µg/ml) Range	50%	90%	% Susceptible/%Intermediate/%Resistant[b] CLSI			EUCAST		
MRSA (1,490)									
Telavancin	≤0.015 — 0.12	0.03	0.06	100.0	-	-[b]	100.0	-	0.0[c]
Clindamycin	≤0.25 — >2	≤0.25	>2	63.7	0.2	36.1	63.5	0.2	36.3
Daptomycin	0.12 — 2	0.25	0.5	99.7	-	-	99.7	-	0.3
Erythromycin	≤0.12 — >16	>16	>16	18.8	2.8	78.4	19.1	0.6	80.3
Gentamicin	≤1 — >8	≤1	>8	87.7	0.4	11.9	87.3	-	12.7
Levofloxacin	≤0.12 — >4	>4	>4	24.9	1.5	73.6	24.9	1.5	73.6
Linezolid	0.25 — 4	1	1	100.0	-	0.0	100.0	-	0.0
Tetracycline	≤0.5 — >8	≤0.5	2	92.3	0.4	7.3	89.6	2.2	8.1
TMP-SMX	≤0.5 — >4	≤0.5	≤0.5	97.7	-	2.3	97.7	0.1	2.1
Vancomycin	0.25 — 4	1	1	99.9	0.1	0.0	99.9	-	0.1
MRSA MDR (569)									
Telavancin	≤0.015 — 0.12	0.06	0.06	100.0	-	-[b]	100.0	-	0.0[c]
Clindamycin	≤0.25 — >2	>2	>2	9.3	0.4	90.3	9.0	0.4	90.7
Daptomycin	0.12 — 2	0.25	0.5	99.3	-	-	99.3	-	0.7
Erythromycin	0.5 — >16	>16	>16	0.5	1.6	97.9	0.5	0.5	98.9
Gentamicin	≤1 — >8	≤1	>8	71.9	0.5	27.6	71.5	-	28.5
Levofloxacin	≤0.12 — >4	>4	>4	0.9	0.5	98.6	0.9	0.5	98.6
Linezolid	0.25 — 4	1	1	100.0	-	0.0	100.0	-	0.0
Tetracycline	≤0.5 — >8	≤0.5	>8	89.1	0.0	10.9	84.0	5.1	10.9
TMP-SMX	≤0.5 — >4	≤0.5	≤0.5	94.6	-	5.4	94.6	0.4	5.1
Vancomycin	0.5 — 4	1	1	99.8	0.2	0.0	99.8	-	0.2
MRSA with vancomycin MIC = 2 - 4 µg/ml (51)									
Telavancin	≤0.015 — 0.12	0.06	0.12	100.0	-	-[b]	100.0	-	0.0[c]
Clindamycin	≤0.25 — >2	>2	>2	25.5	0.0	74.5	25.5	0.0	74.5
Daptomycin	0.25 — 2	0.5	1	98.0	-	-	98.0	-	2.0
Erythromycin	≤0.12 — >16	>16	>16	9.8	2.0	88.2	9.8	2.0	88.2
Gentamicin	≤1 — >8	≤1	>8	72.5	0.0	27.5	72.5	-	27.5
Levofloxacin	≤0.12 — >4	>4	>4	13.7	0.0	86.3	13.7	0.0	86.3
Linezolid	0.25 — 2	1	2	100.0	-	0.0	100.0	-	0.0
Tetracycline	≤0.5 — >8	≤0.5	2	90.2	0.0	9.8	84.3	5.9	9.8
TMP-SMX	≤0.5 — >4	≤0.5	≤0.5	94.1	-	5.9	94.1	0.0	5.9
Vancomycin	2 — 4	2	2	98.0	2.0	0.0	98.0	-	2.0

a. MRSA = methicillin-resistant *S. aureus*; TMP-SMX = trimethoprim-sulfamethoxazole; MDR = multidrug resistance (defined as MRSA resistant to three or more drug classes in addition to β-lactam agents).
b. Breakpoint criteria for telavancin according to CLSI (M100-S26, 2016) and EUCAST breakpoint criteria for telavancin comparator agents, as available.
c. - = Breakpoint not available.

Figure 1: Telavancin, daptomycin and vancomycin MIC distributions against all MRSA causing bacteremia. Data presented as the cumulative percentage of isolates inhibited at each MIC (µg/ml). MIC_{50} differences between drugs are depicted.

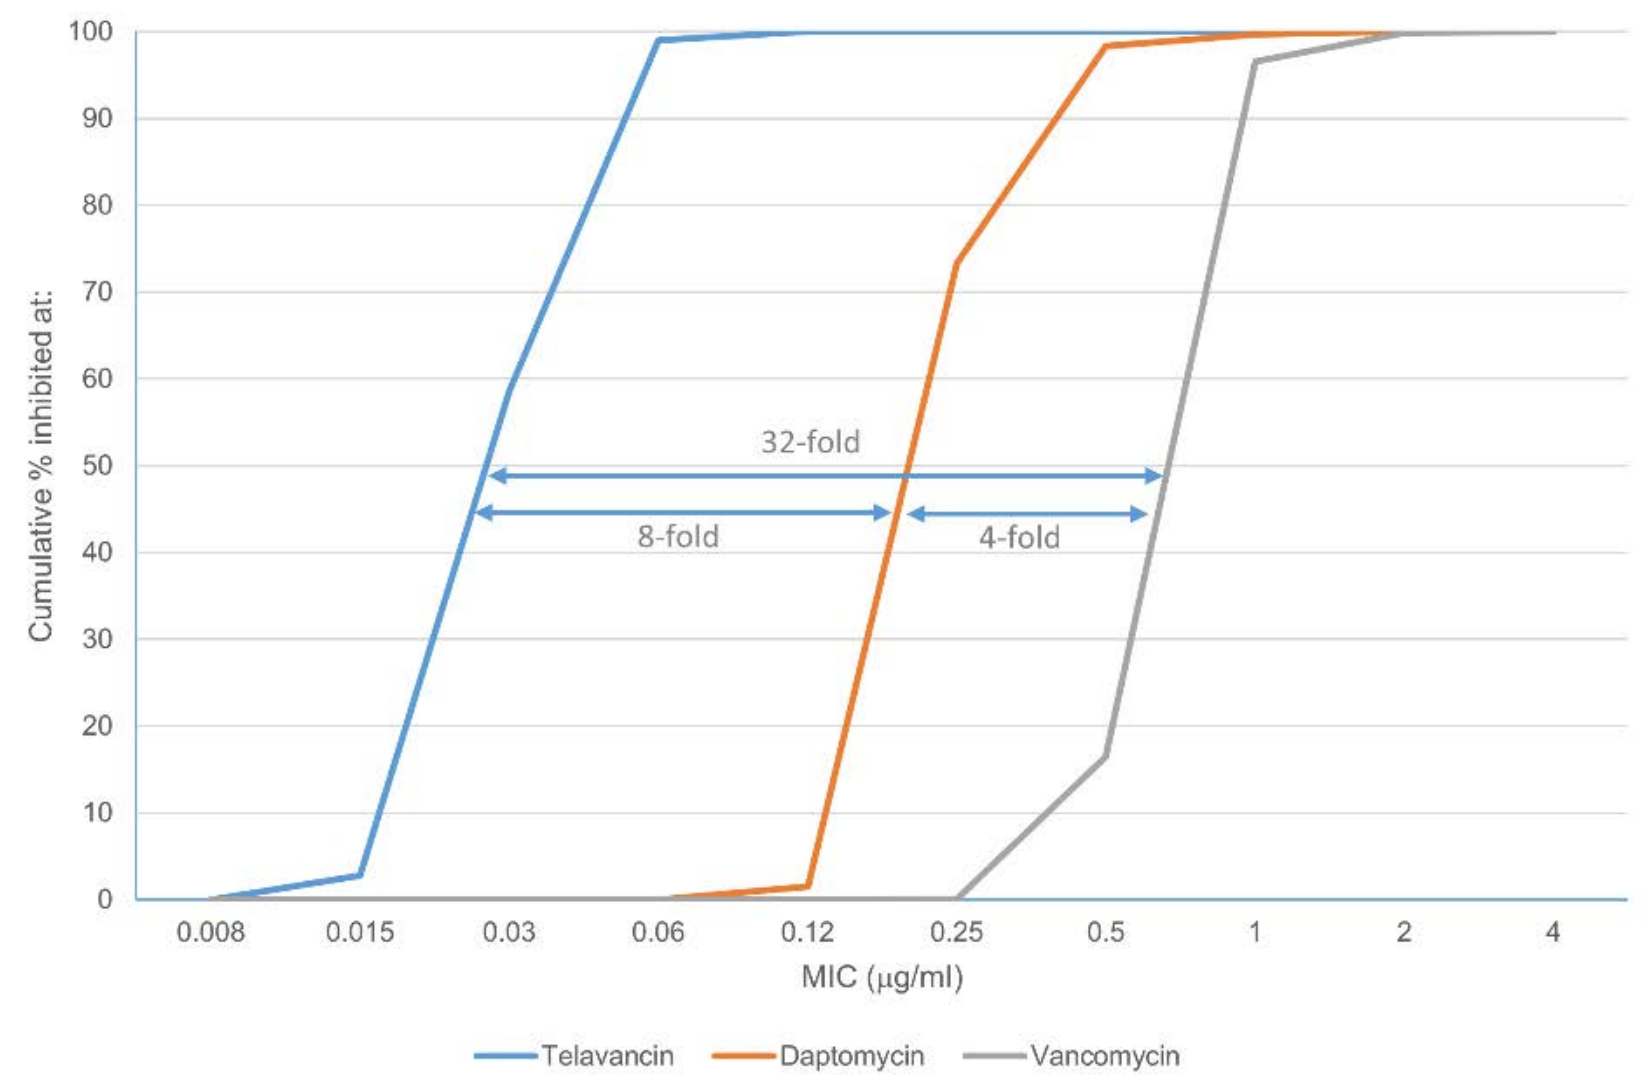
100
90
80
70
60
50
40
30
20
10
0
Cumulative % inhibited at:
32-fold
8-fold
4-fold
0.008
0.015
0.03
0.06
0.12
0.25
0.5
1
2
4
MIC (µg/ml)
Telavancin
Daptomycin
Vancomycin

CONCLUSIONS

- Telavancin (100.0% susceptible) demonstrated potent *in vitro* activity against this global and contemporary collection of *S. aureus* causing bacteremia, including against resistant subsets and isolates causing endocarditis.
- In addition, telavancin had *in vitro* potency at least 4-fold greater than other comparator antimicrobial agents (daptomycin and vancomycin) recommended by the current guidelines for the treatment of bacteremia caused by MRSA and MDR subsets.
- These *in vitro* results support further investigation of telavancin as a candidate for the treatment of bacteremia caused by *S. aureus* and resistant subsets, including those isolates responsible for endocarditis.

ACKNOWLEDGEMENTS

The research and publication process was supported by Theravance Biopharma Antibiotics, Inc. Poster production support was coordinated by AlphaBioCom, LLC. funded by Theravance Biopharma Antibiotics, Inc.

REFERENCES

1. Clinical and Laboratory Standards Institute (2015). *M07-A10. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; Approved Standard - Tenth edition*. Wayne, PA, USA.
2. Clinical and Laboratory Standards Institute (2016). *M100-S26. Performance standards for antimicrobial susceptibility testing: 26th Informational Supplement*. Wayne, PA, USA.
3. Corey GR, Rubinstein E, Stryjewski ME, Bassetti M, Barriere SL (2015). Potential role for telavancin in bacteremic infections due to gram-positive pathogens: focus on *Staphylococcus aureus*. *Clin Infect Dis*. 60: 787-796.
4. EUCAST (2016). Breakpoint tables for interpretation of MICs and zone diameters. Version 6.0, January 2016. Available at: http://www.eucast.org/clinical_breakpoints/. Accessed January 2016.
5. Kreisel K, Roghmann MC, Shardell M, Stine OC, Perencevich E, Lesse A, Gordin F, Climo M, Johnson JK (2010). Assessment of the 48-hour rule for identifying community-associated methicillin-resistant *Staphylococcus aureus* infection complicated by bacteremia. *Infect Control Hosp Epidemiol* 31: 657-659.
6. Laupland KB, Lyytikainen O, Sogaard M, Kennedy KJ, Knudsen JD, Ostergaard C, Galbraith JC, Valiquette L, Jacobsson G, Collignon P, Schonheyder HC, International Bacteremia Surveillance Collaborative (2013). The changing epidemiology of *Staphylococcus aureus* bloodstream infection: a multinational population-based surveillance study. *Clin Microbiol Infect* 19: 465–471.
7. Mendes RE, Sader HS, Flamm RK, Farrell DJ, Jones RN (2015). Telavancin *in vitro* activity against a collection of methicillin-resistant *Staphylococcus aureus* isolates, including resistant subsets, from the United States. *Antimicrob Agents Chemother* 59: 1811-1814.
8. Stryjewski ME, Corey GR (2014). Methicillin-resistant *Staphylococcus aureus*: An evolving pathogen. *Clin Infect Dis* 58: S10–S19.
9. Tong SY, Davis JS, Eichenberger E, Holland TL, Fowler VG Jr (2015). *Staphylococcus aureus* infections: epidemiology, pathophysiology, clinical manifestations, and management. *Clin Microbiol Rev*. 28: 603-661.
10. VanEperen AS, Segreti J (2016). Empirical therapy in Methicillin-resistant *Staphylococcus aureus* infections: An Up-To-Date approach. *J Infect Chemother* 8: S1341-321.

Presented at the American Society for Microbiology Microbe; June 16 – 20, 2016; Boston, Massachusetts.