Antimicrobial Activity of Ceftazidime-Avibactam and Comparator Agents against *Pseudomonas aeruginosa* and *Enterobacterales* Causing Pneumonia in Cystic Fibrosis Patients

LR Duncan, JM Streit, M Castanheira, HS Sader

JMI Laboratories, North Liberty, IA, USA

NACFC 2020

Helio S. Sader, MD, PhD, FIDSA
JMI Laboratories
345 Beaver Kreek Centre, Suite A
North Liberty, Iowa 52317
Phone: (319) 665-3370
Fax: (319) 665-3371
Email: helio-sader@jmilabs.com

INTRODUCTION

- More than 50% of cystic fibrosis (CF) individuals aged 18 years and older in the USA are infected with *Pseudomonas aeruginosa*, of which approximately one-third is multidrug-resistant (MDR).
- Very few agents remain active against MDR *P. aeruginosa* and rapidly introducing appropriate antimicrobial therapy is crucial to reduce morbidity and mortality of patients with infections caused by these organisms.
- Ceftazidime-avibactam is approved by the United States Food and Drug Administration (US FDA) and the European Medicines Agency (EMA) to treat hospital-acquired bacterial pneumonia, including ventilator-associated bacterial pneumonia (HABP/VABP), complicated intra-abdominal infections (cIAIs) in combination with metronidazole, and complicated urinary tract infections (UTIs), including pyelonephritis.
- We evaluated the antimicrobial susceptibility patterns of *P. aeruginosa* and *Enterobacterales* isolated from CF patients with pneumonia.

MATERIALS AND METHODS

- Bacterial isolates determined to be significant by local criteria as the reported probable cause of respiratory infection were consecutively collected from CF patients (1/patient).
- Isolates were collected from 43 medical centers worldwide (19 nations) in 2018–2019.
- The organism collection included 273 *P. aeruginosa* and 62 *Enterobacterales* isolates.
- Susceptibility testing was performed by reference broth microdilution methods at a central laboratory (JMI Laboratories, North Liberty, Iowa).
- Antimicrobial agents tested include ceftazidime-avibactam (CAZ-AVI), ceftolozane-tazobactam (C-T), piperacillin-tazobactam (PIP-TAZ), meropenem (MEM), ceftazidime (CAZ), ceftriaxone (CRO; *Enterobacterales* only), levofloxacin (LEV), tobramycin (TOB), and gentamicin (GEN).
- MDR was defined as nonsusceptibility (CLSI breakpoints) to at least 1 drug in 3 classes.

RESULTS

- Ceftazidime-avibactam (MIC$_{50/90}$, 2/8 mg/L; 96.0% susceptible [S]) was the most active agent against *P. aeruginosa*, followed by ceftolozane-tazobactam (MIC$_{50/90}$, 1/4 mg/L; 90.5%S), ceftazidime (MIC$_{50/90}$, 2/>32 mg/L; 80.6%S), piperacillin-tazobactam (MIC$_{50/90}$, 4/128 mg/L; 80.2%S), and tobramycin (MIC$_{50/90}$, 2/>16 mg/L; 76.6%S; Figure 1).
- Ceftazidime-avibactam retained activity against *P. aeruginosa* isolates nonsusceptible to meropenem (86.5%S to ceftazidime-avibactam), piperacillin-tazobactam (85.2%S to ceftazidime-avibactam), or ceftazidime (79.2%S to ceftazidime-avibactam; Table 1 and Figure 2).
- Moreover, 65.4% of ceftolozane-tazobactam-nonsusceptible isolates remained susceptible to ceftazidime-avibactam (Table 1).
- Among *P. aeruginosa*, 36.3% were multidrug-resistant (MDR, resistant to ≥3 classes), and 88.9% and 73.7% of MDR *P. aeruginosa* were susceptible to ceftazidime-avibactam and ceftolozane-tazobactam, respectively (Figure 2).
- Among *P. aeruginosa* isolates nonsusceptible to piperacillin-tazobactam, meropenem, and ceftazidime (13.9% of total), susceptibility to ceftazidime-avibactam and ceftolozane-tazobactam was 78.9% and 47.4%, respectively (Figure 2).
- The most active agents against *Enterobacterales* were ceftazidime-avibactam (MIC$_{50/90}$, 0.12/0.5 mg/L; 100.0%S) and meropenem (MIC$_{50/90}$, 0.03/0.12 mg/L; 96.8%S; Figure 3).
- Ceftolozane-tazobactam (MIC$_{50/90}$, 0.25/4 mg/L) was active against 88.7% of ENT.
- Among *Enterobacterales* isolates, 17.7% were MDR.

Table 1. Cross-resistance among β-lactams and β-lactamase inhibitor combinations when tested against *P. aeruginosa* isolates from CF patients (2018–2019)

Antimicrobial	% Susceptible by resistant subset (no. of isolates)				
	MEM-NS (74)	PIP-TAZ-NS (54)	CAZ-NS (53)	C-T-NS (26)	CAZ-AVI-NS (11)
MEM	0.0	20.4	17.0	3.8	9.1
PIP-TAZ	41.9	0.0	15.1	3.8	27.2
CAZ	40.5	16.7	0.0	3.8	0.0
C-T	66.2	63.0	52.8	0.0	18.2
CAZ-AVI	86.5	85.2	79.2	65.4	0.0

Abbreviations: CAZ, ceftazidime; MEM, meropenem; PIP-TAZ, piperacillin-tazobactam; C-T, ceftolozane-tazobactam; AVI, avibactam; NS, not susceptible.

CONCLUSIONS

- Ceftazidime-avibactam exhibited potent and broad-spectrum activity against *P. aeruginosa* (96.0%S) and *Enterobacterales* (100.0%S) isolated from CF patients worldwide and retained good activity against isolates resistant to other antimicrobials, including MDR organisms.
- Ceftazidime-avibactam and ceftolozane-tazobactam exhibited similar *P. aeruginosa* coverage, but ceftazidime-avibactam was more active than ceftolozane-tazobactam against *P. aeruginosa* resistant subsets.
- Ceftazidime-avibactam represents a valuable option to treat CF patients with respiratory tract infections.

ACKNOWLEDGEMENTS

This study was supported by Allergan (prior to its acquisition by AbbVie). Allergan (now AbbVie) was involved in the design and decision to present these results and JMI Laboratories received compensation fees for services in relation to preparing the manuscript. Allergan (now AbbVie) had no involvement in the collection, analysis, and interpretation of data.

REFERENCES

1. Atkin SD, Abid S, Foster M, Bose M, Keller A, Hollaway R, Sader HS, Greenberg DE, Finklea JD, Castanheira M, Jain R (2018). Multidrug-resistant *Pseudomonas aeruginosa* from sputum of patients with cystic fibrosis demonstrates a high rate of susceptibility to ceftazidime-avibactam. *Infect Drug Resist* 17:1499–1510.
2. AVYCAZ® (2019). Avycaz (ceftazidime-avibactam) package insert. Allergan USA, Inc. Available at https://www.allergan.com/assets/pdf/avycaz_pi.
3. Clinical and Laboratory Standards Institute (2018). *M07Ed11E. Methods for dilution antimicrobial susceptibility tests for bacteria that grow aerobically; approved standard: eleventh edition*. Wayne, PA: CLSI.
4. Cystic Fibrosis Foundation Patient Registry (2018) Annual Data Report. Available at https://www.cff.org/Research/Researcher-Resources/Patient-Registry/2018-Patient-Registry-Annual-Data-Report.pdf.
5. Sader HS, Flamm RK, Carvalhaes CG, Castanheira M (2020). Comparison of ceftazidime-avibactam and ceftolozane-tazobactam in vitro activities when tested against gram-negative bacteria isolated from patients hospitalized with pneumonia in United States medical centers (2017–2018). *Diagn Microbiol Infect Dis* 96:114833.

Figure 1. Antimicrobial susceptibility of *P. aeruginosa* (n=273) collected from cystic fibrosis patients worldwide (2018–2019)

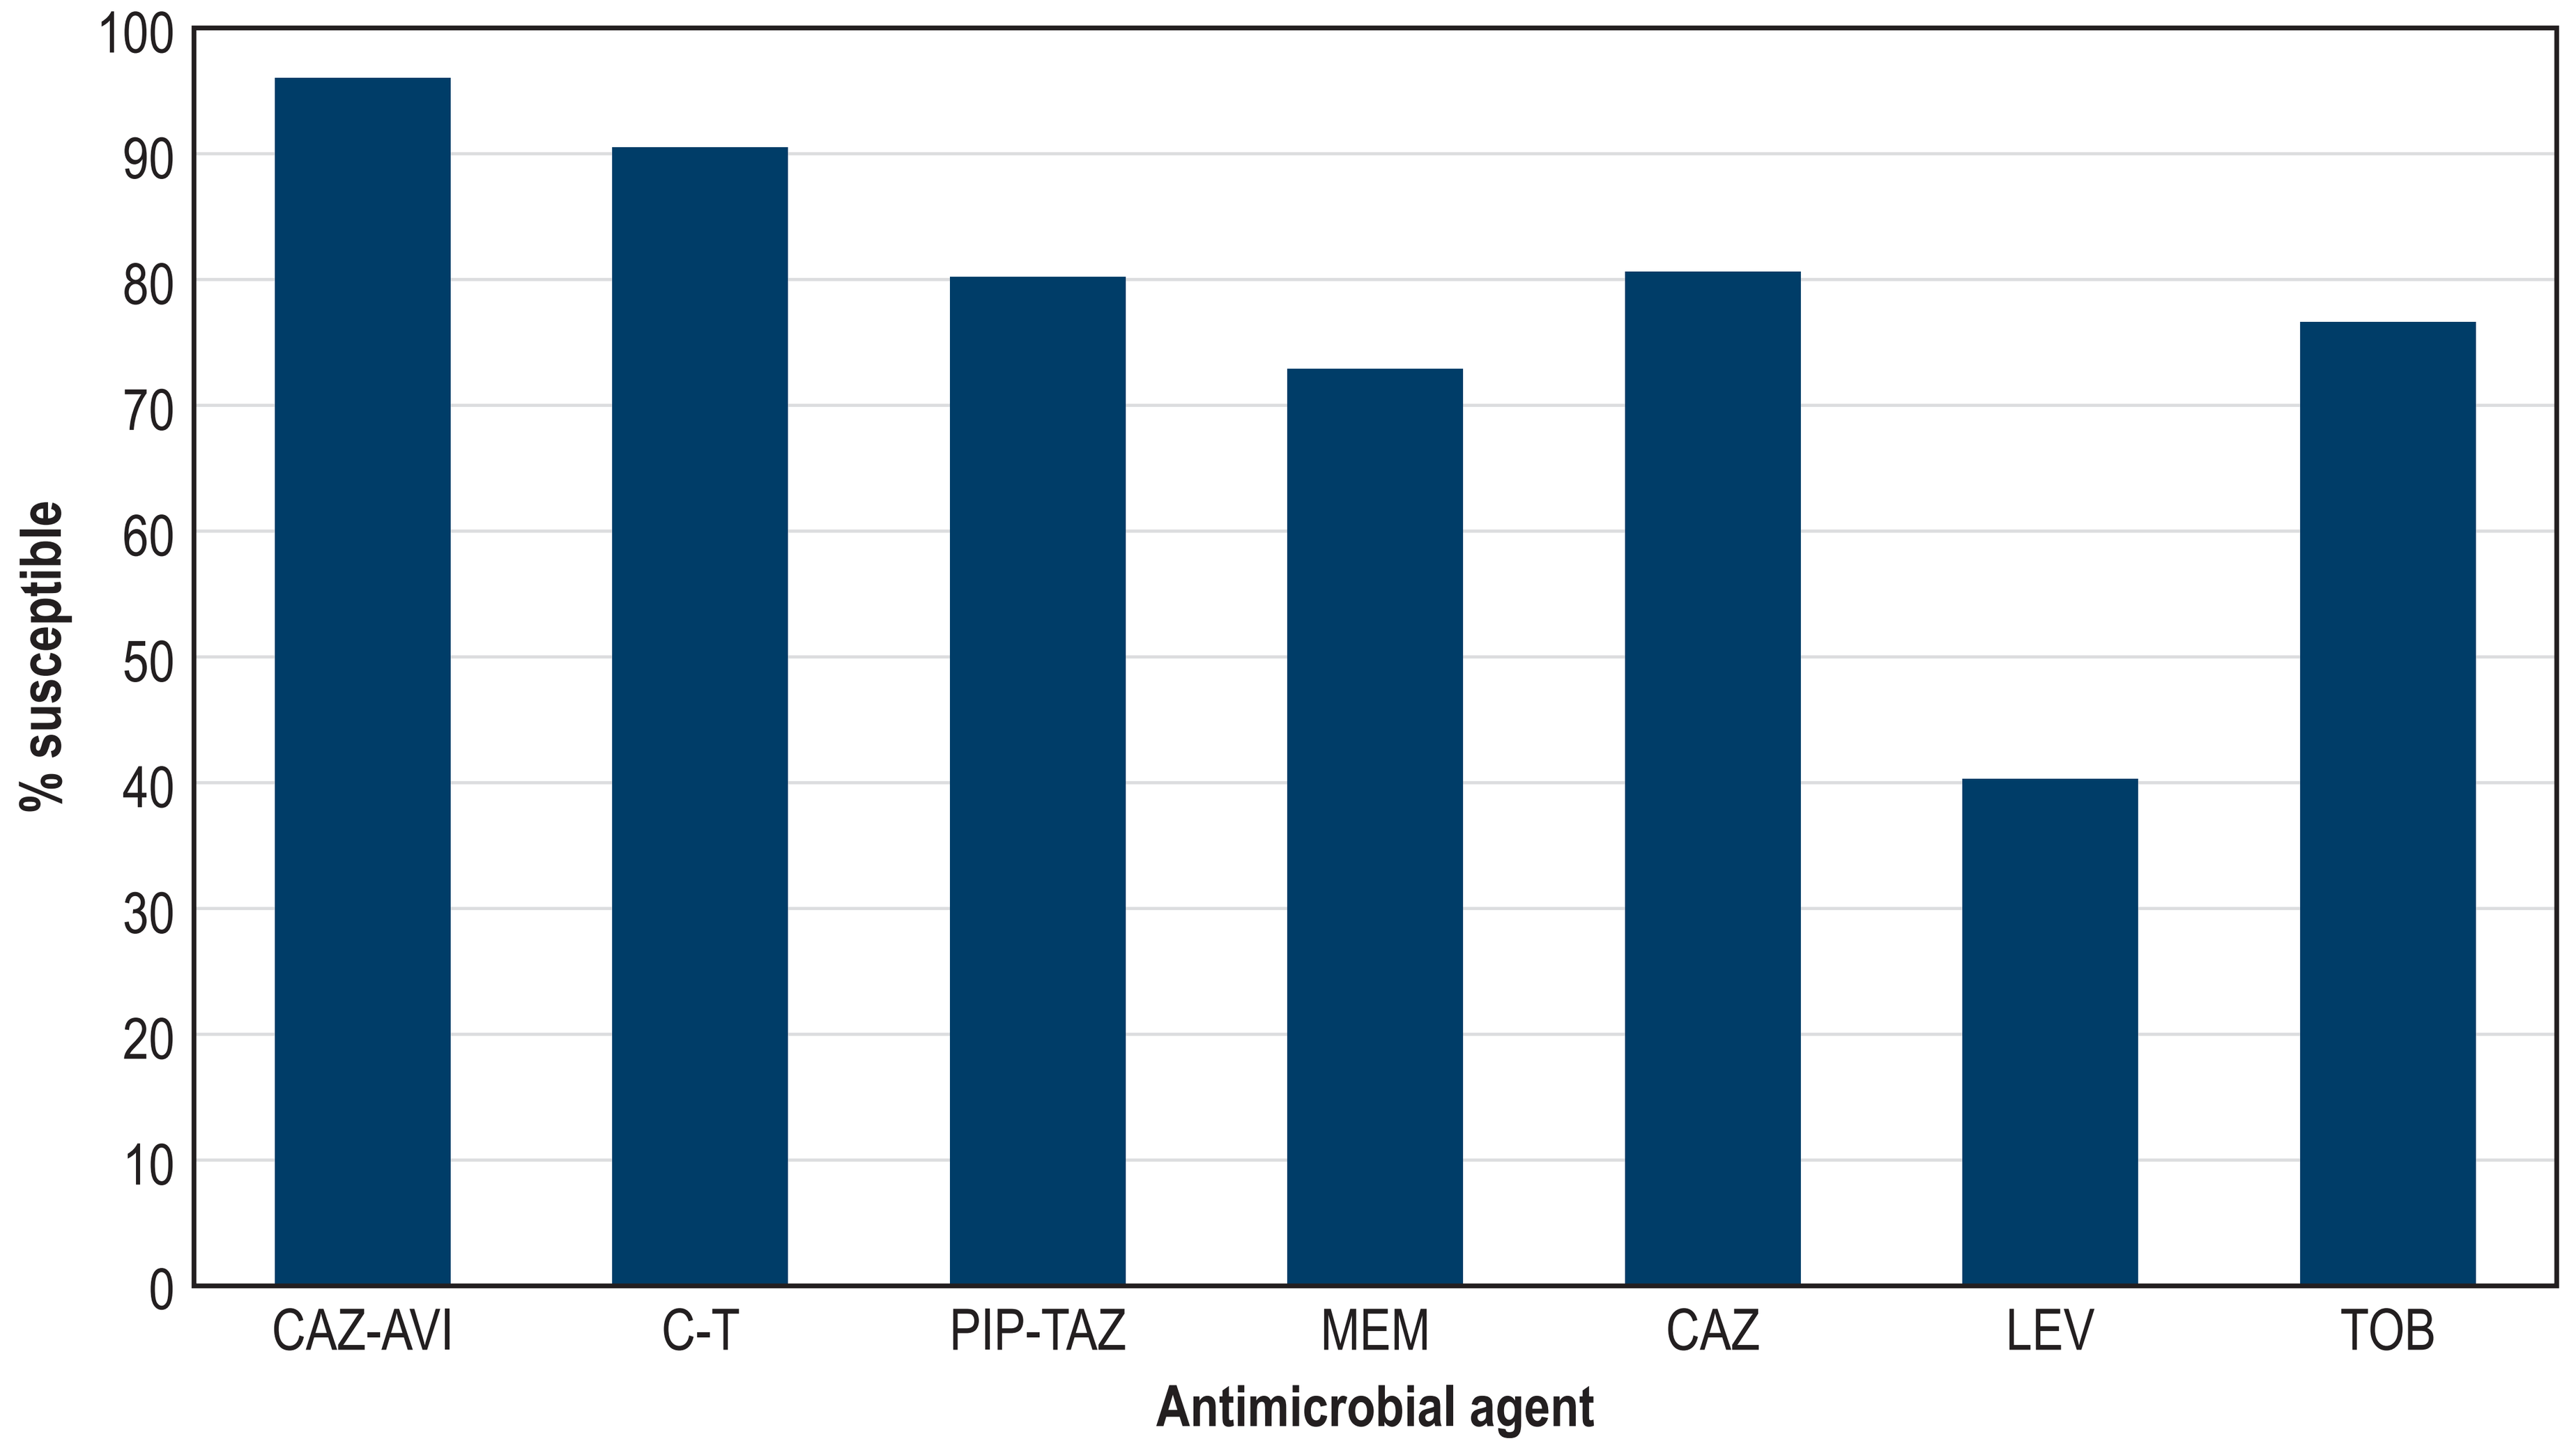
% susceptible
0
10
20
30
40
50
60
70
80
90
100
CAZ-AVI
C-T
PIP-TAZ
MEM
CAZ
LEV
TOB
Antimicrobial agent

Abbreviations: CAZ-AVI, ceftazidime-avibactam; C-T, ceftolozane-tazobactam; PIP-TAZ, piperacillin-tazobactam; MEM, meropenem; CAZ, ceftazidime; LEV, levofloxacin; TOB, tobramycin.

Figure 2. Antimicrobial activity of ceftazidime-avibactam (CAZ-AVI), ceftolozane-tazobactam (C-T), and tobramycin (TOB) tested against *P. aeruginosa* resistant subsets from cystic fibrosis patients (2018–2019)

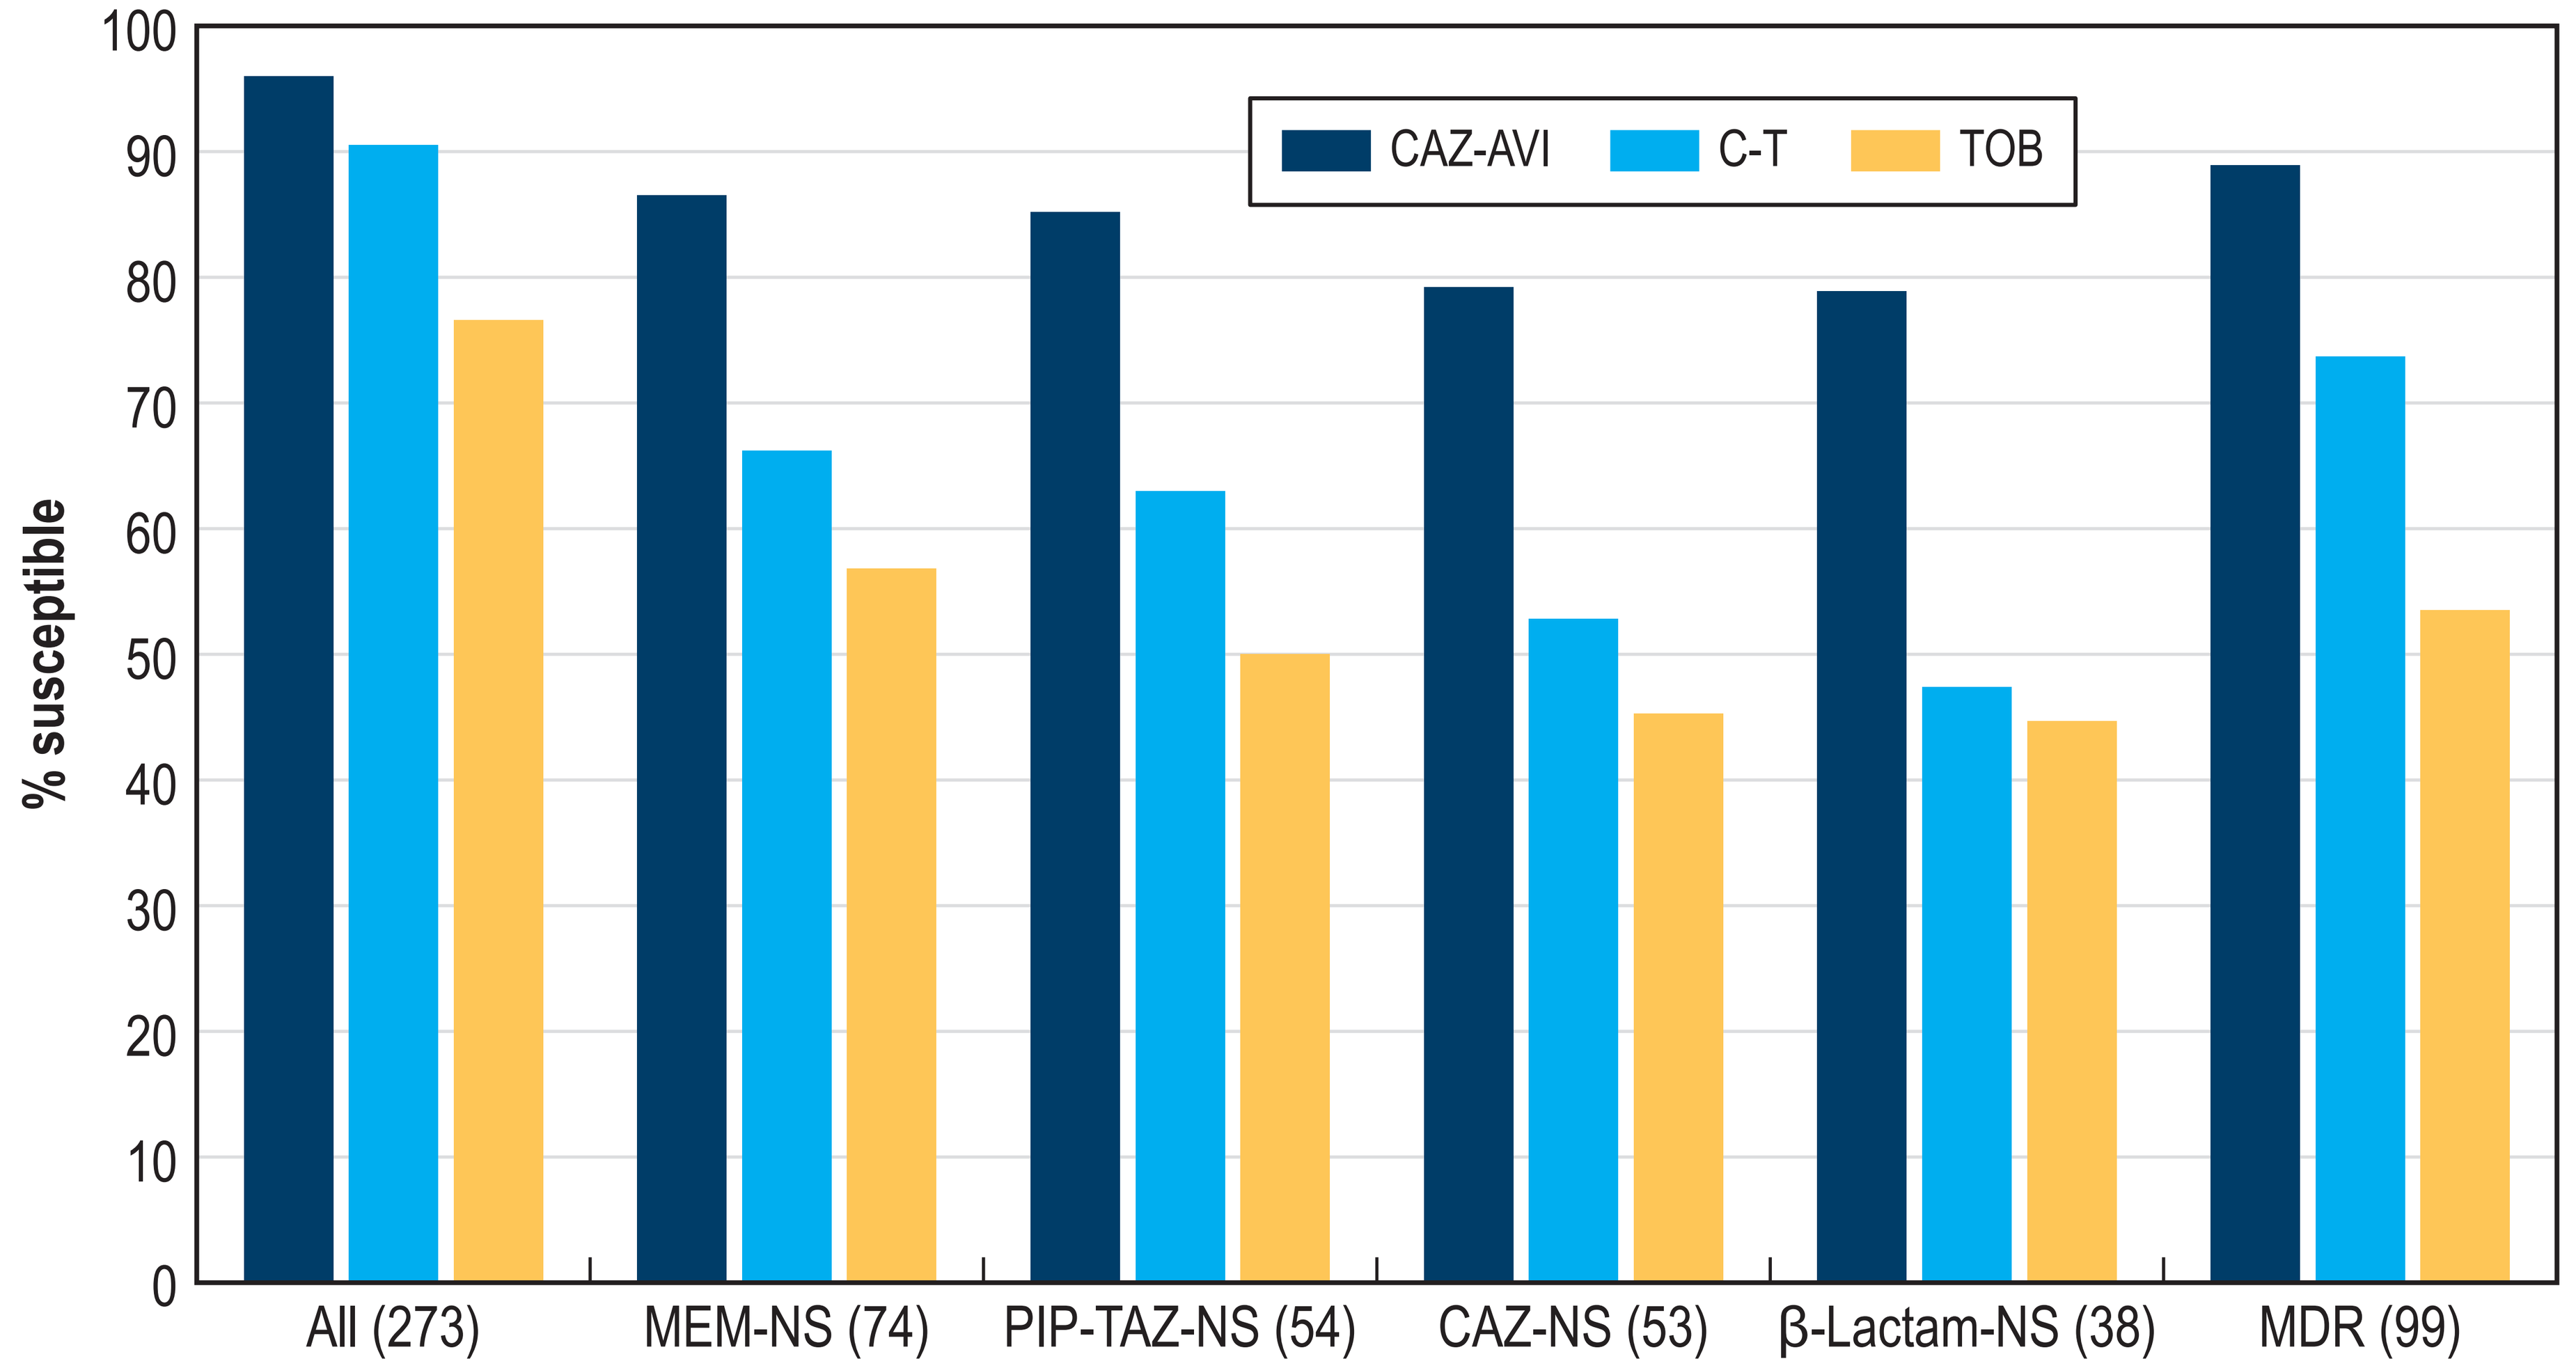
% susceptible
0
10
20
30
40
50
60
70
80
90
100
CAZ-AVI
C-T
TOB
All (273)
MEM-NS (74)
PIP-TAZ-NS (54)
CAZ-NS (53)
β-Lactam-NS (38)
MDR (99)

Abbreviations: MEM, meropenem; NS, nonsusceptible; PIP-TAZ, piperacillin-tazobactam; CAZ, ceftazidime; β-lactam-NS, isolates not susceptible to meropenem, piperacillin-tazobactam, and ceftazidime; and MDR, multidrug-resistant.

Figure 3. Antimicrobial susceptibility of *Enterobacterales* (n=62) collected from cystic fibrosis patients worldwide (2018–2019)

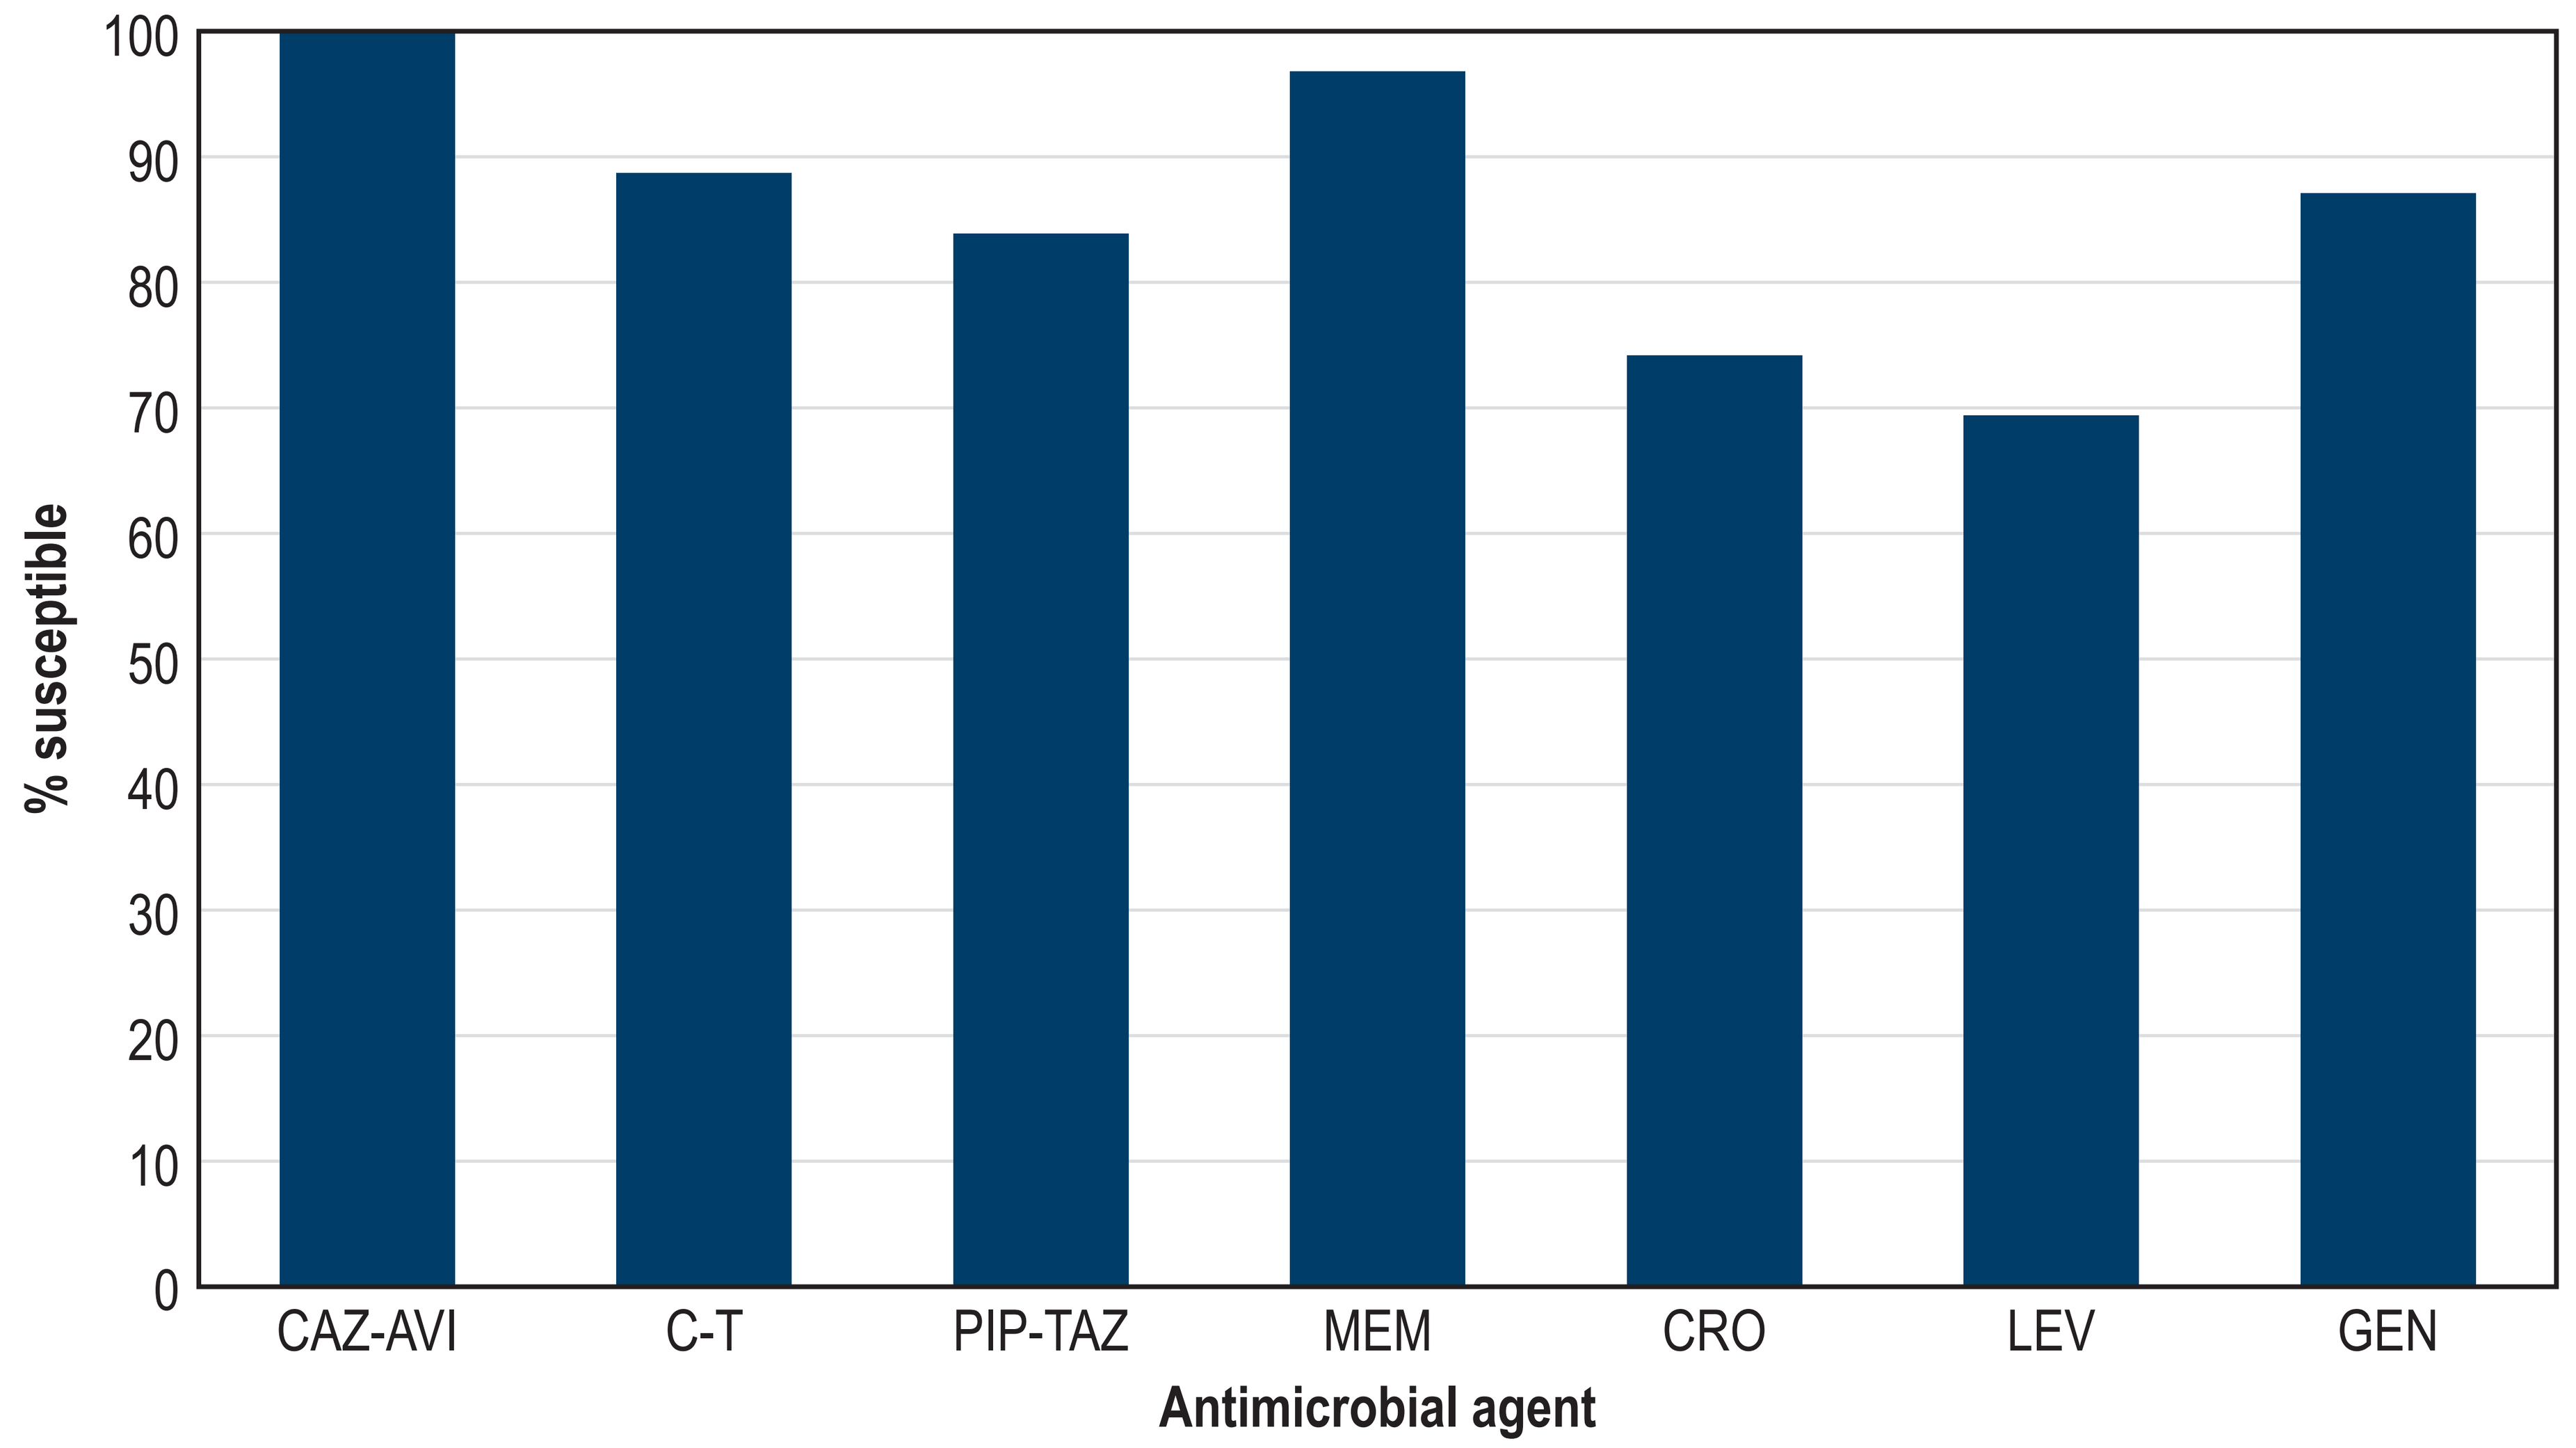
% susceptible
0
10
20
30
40
50
60
70
80
90
100
CAZ-AVI
C-T
PIP-TAZ
MEM
CRO
LEV
GEN
Antimicrobial agent

Abbreviations: CAZ-AVI, ceftazidime-avibactam; C-T, ceftolozane-tazobactam; PIP-TAZ, piperacillin-tazobactam; MEM, meropenem; CRO, ceftriaxone; LEV, levofloxacin; GEN, gentamicin.

abbvie